Predictive Markers of Response to the MDM2 Degrader KT-253

Nancy Dumont[1]*****, Rahul Karnik[1]*, Yogesh Chutake[1], Joyoti Dey[1], Susanne Breitkopf[1], Charles Lincoln Howarth[1], Carol Meeske[1], Alyssa Fasciano[1], Samson Ghilu[2], Peter Houghton[2], Rebecca Mosher[1], Jared Gollob[1], and Juliet Williams[1]
*These authors contributed equally to this work.

[1]Kymera Therapeutics, Watertown, Massachusetts, USA.
[2]Greehey Children's Cancer Research Institute, University of Texas Health Science Center at San Antonio, San Antonio, Texas, USA.

INTRODUCTION

Murine double minute 2 (MDM2) is an E3 ligase that regulates the tumor suppressor p53. Clinical trials employing small-molecule MDM2/p53 interaction inhibitors (SMIs) have demonstrated limited activity, underscoring an unmet need for a better approach to target MDM2.

We have developed a highly potent and selective heterobifunctional degrader of MDM2, KT-253, that overcomes the MDM2 feedback loop seen with SMIs and induces acute apoptosis in a range of hematologic and solid tumor lines. KT-253 is being evaluated in a Phase 1 clinical trial in adult patients with advanced solid tumors, lymphoma, myelofibrosis, high-grade myeloid malignancies and acute lymphocytic leukemias (NCT05775406).

The objective of this work was to identify a patient selection strategy that enriched for tumor types where KT-253 induces an acute apoptotic response and robust antitumor activity with Q3W dosing. We incorporated our understanding of biologic mechanisms, *in vitro* and *in vivo* studies, expression profiling, and machine learning to define biomarkers associated with response.

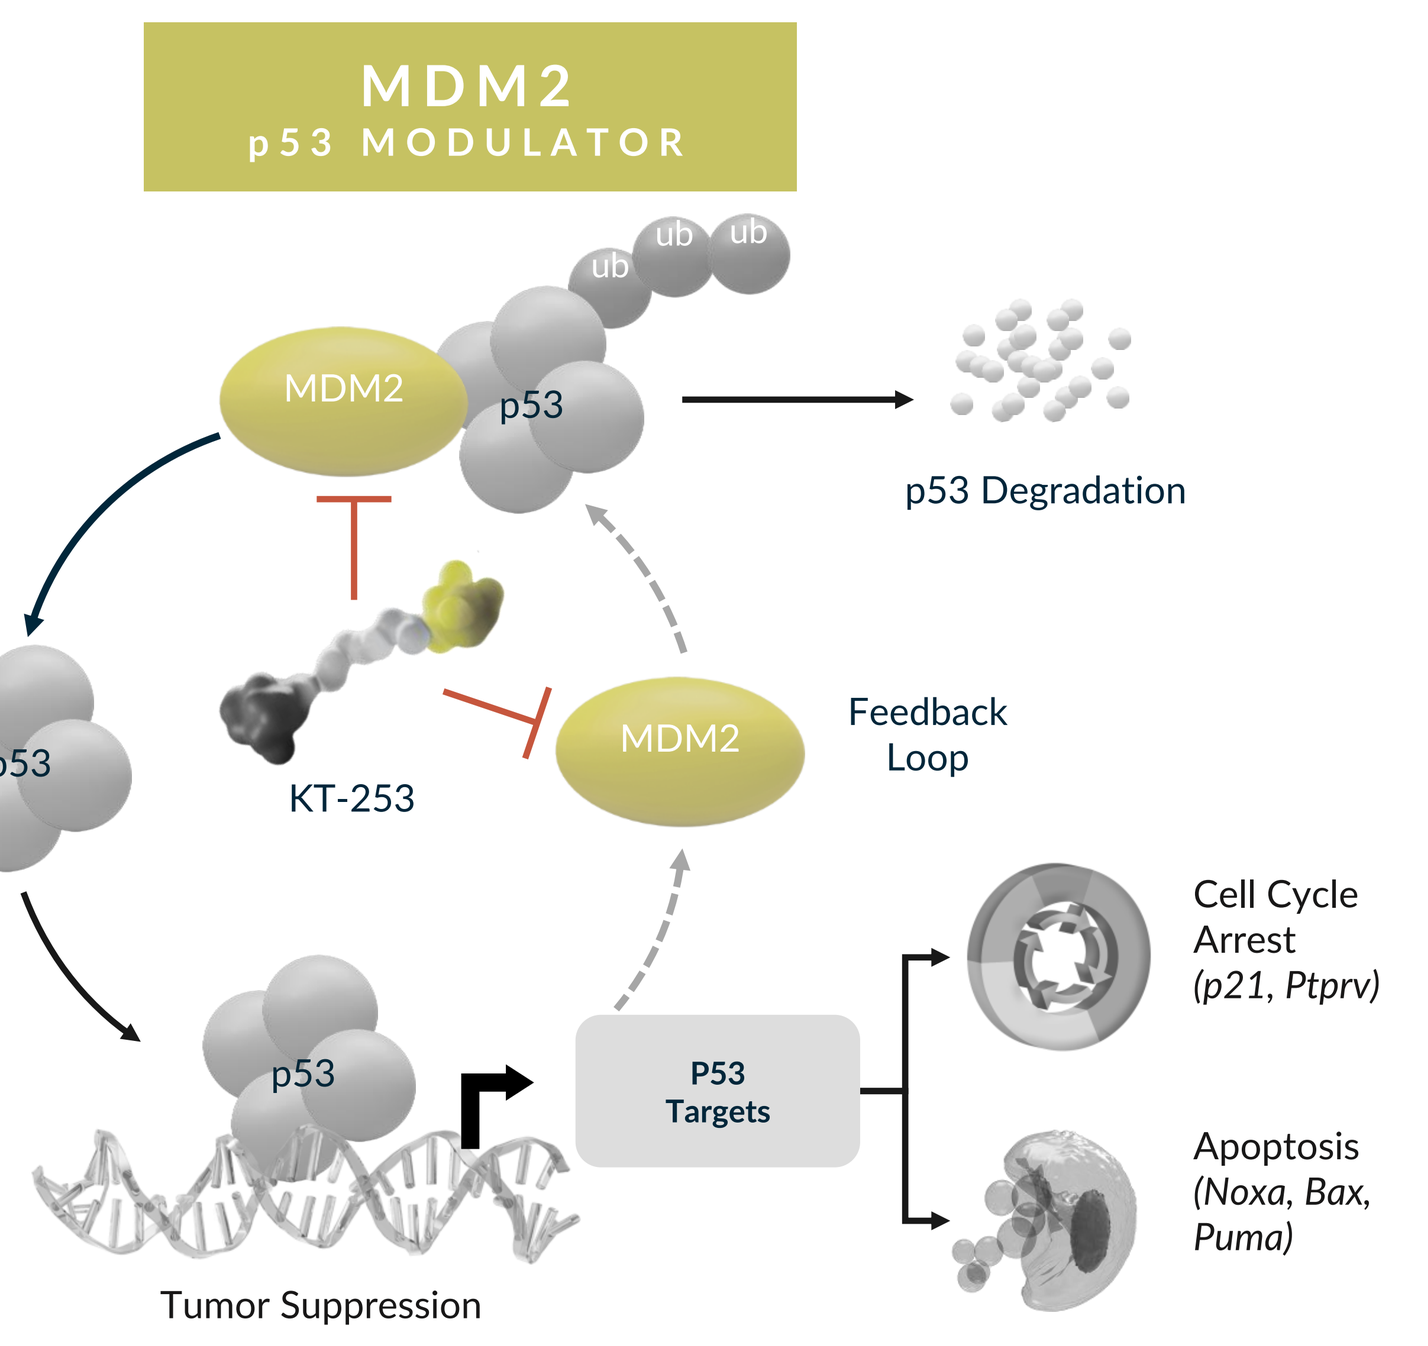
MDM2
p53 MODULATOR
MDM2
p53
ub
p53 Degradation
p53
KT-253
MDM2
Feedback Loop
Cell Cycle Arrest (p21, Ptprv)
p53
P53 Targets
Apoptosis (Noxa, Bax, Puma)
Tumor Suppression

METHODS

We explored the activity of KT-253 *in vitro* in a panel of 350 p53 wild-type cell lines spanning 48 indications. The combination of both cell growth inhibition and apoptosis was evaluated using CTG (Promega) and Caspase-Glo 3/7 assays in 187 lines. Twenty-four pediatric patient-derived xenograft (PDX) models representing 6 tumor types (Ewing sarcoma, rhabdomyosarcoma, rhabdoid tumor, Wilms tumor, hepatoblastoma, and neuroblastoma) and 101 adult PDX models representing 5 solid tumor types (breast, lung, colorectal, gastric, melanoma) were evaluated using a single mouse trial design. KT-253 was administered intravenously every 3 weeks (Q3W). Baseline expression profiles of the adult PDX models were used in a machine learning framework to build a predictive signature of response to KT-253. The signature was validated in 19 prospectively selected PDX models.

RESULTS

Figure 1: KT-253 is Active in Multiple p53 WT Hematological and Solid Tumor Cell Lines *In Vitro*

A) Heme Cell Lines (n=68)

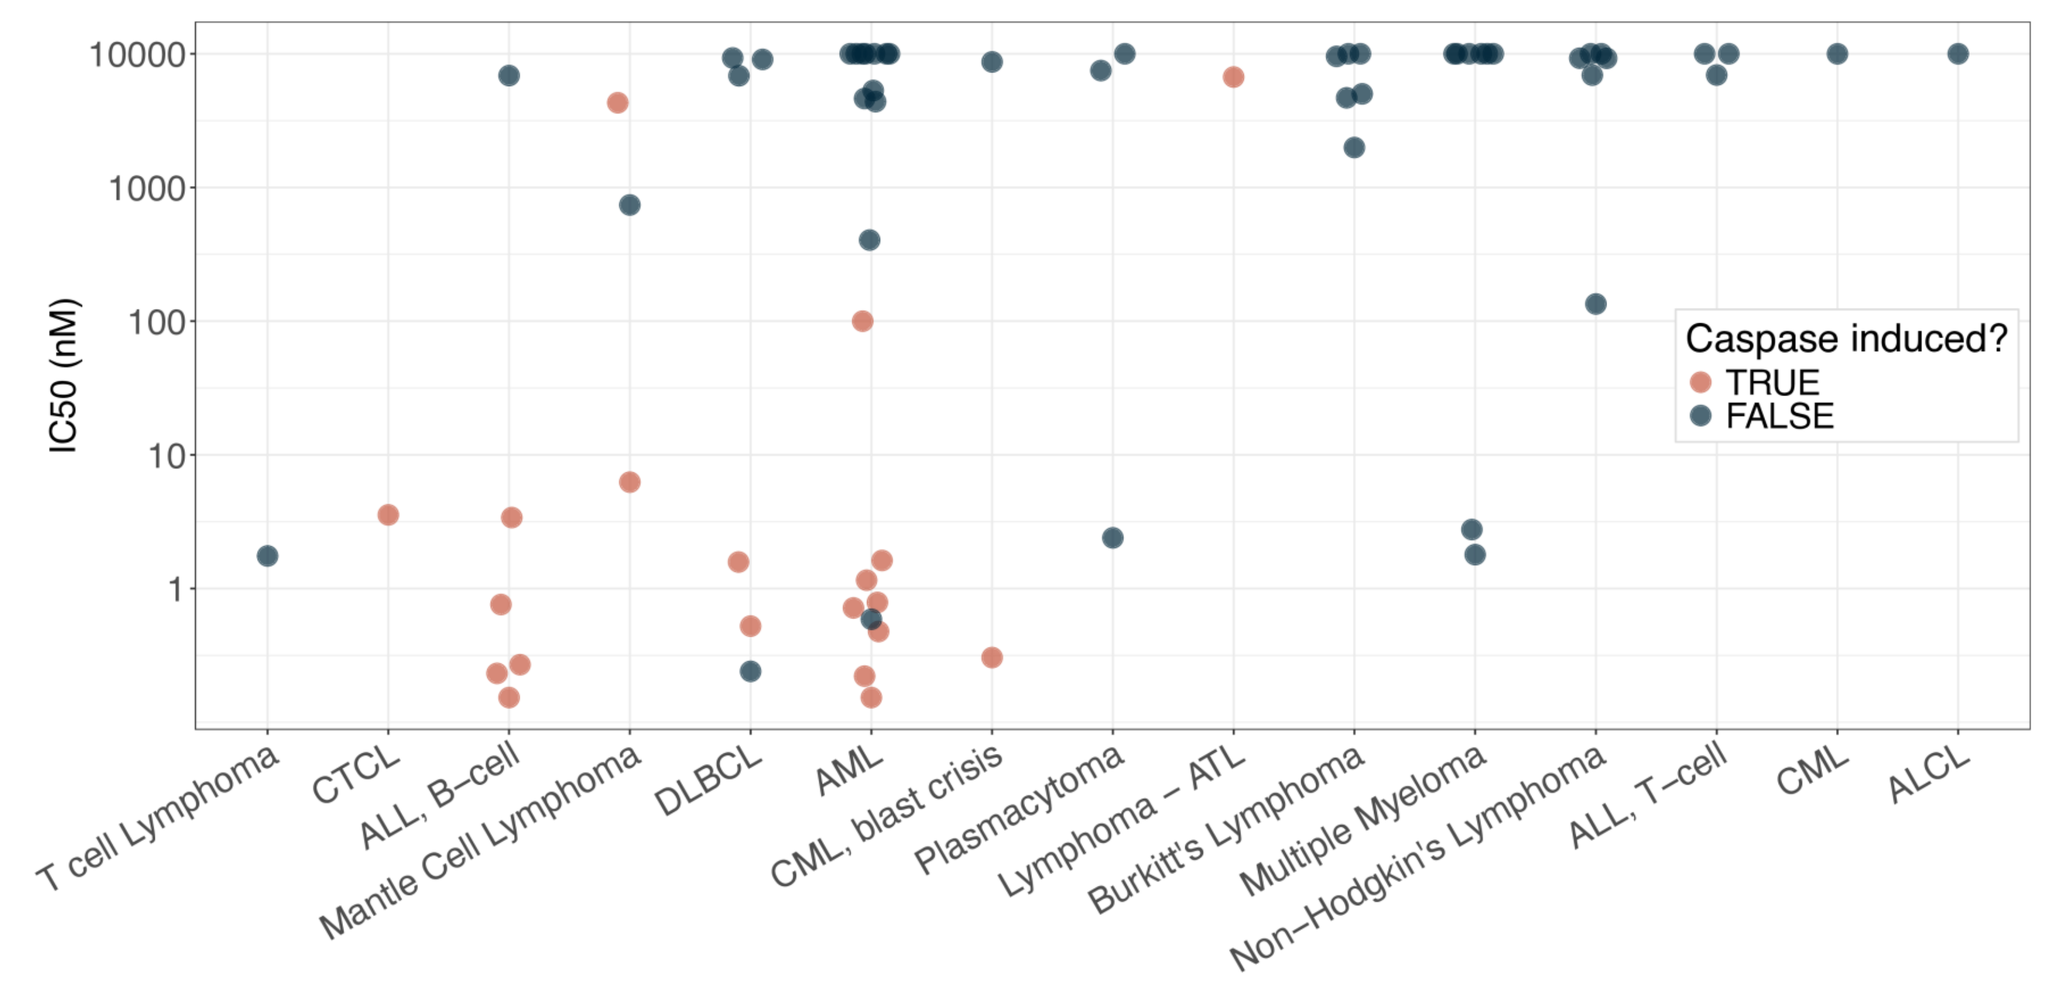
IC50 (nM)
Caspase induced?
TRUE
FALSE
T cell Lymphoma
CTCL
ALL, B-cell
Mantle Cell Lymphoma
DLBCL
AML
CML, blast crisis
Plasmacytoma
Lymphoma – ATL
Burkitt's Lymphoma
Multiple Myeloma
Non–Hodgkin's Lymphoma
ALL, T-cell
CML
ALCL

B) Solid Tumor Cell Lines (n=119)

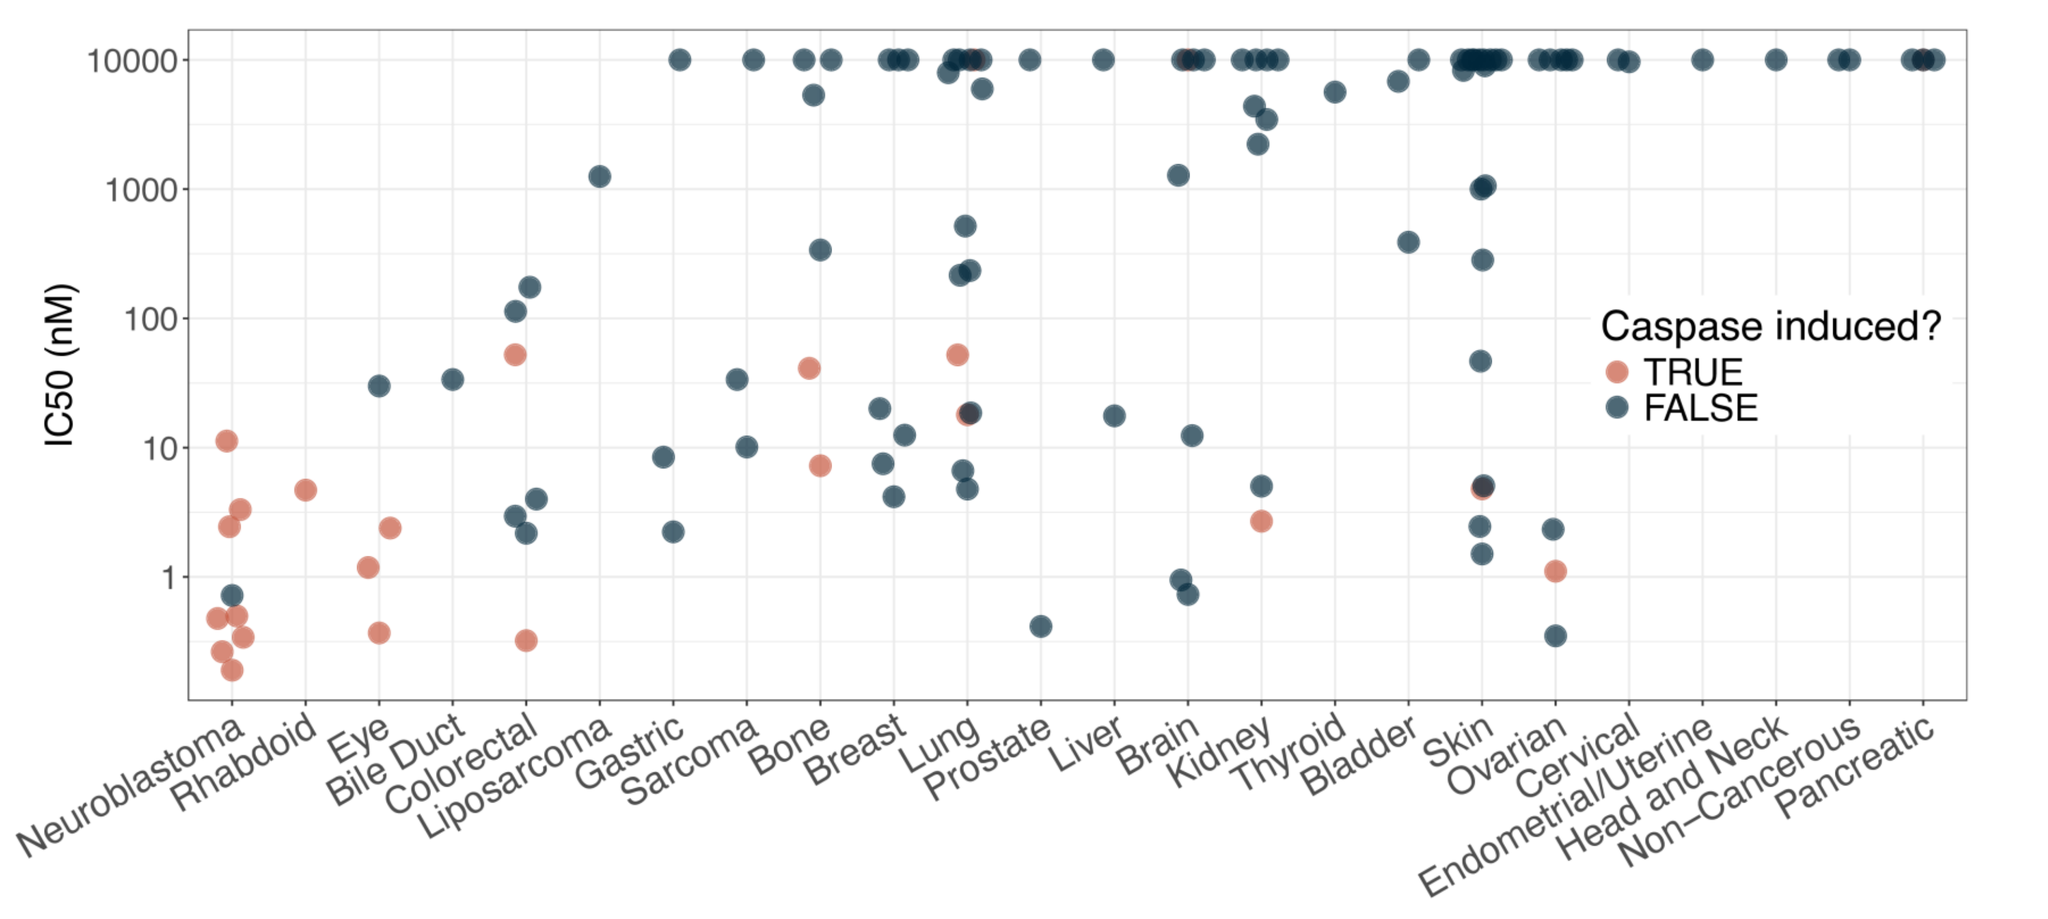
IC50 (nM)
Caspase induced?
TRUE
FALSE
Neuroblastoma
Rhabdoid
Eye
Bile Duct
Colorectal
Liposarcoma
Gastric
Sarcoma
Bone
Breast
Lung
Prostate
Liver
Brain
Kidney
Thyroid
Bladder
Skin
Ovarian
Cervical
Endometrial/Uterine
Head and Neck
Non-Cancerous
Pancreatic

In vitro screening indicates a wide range of A) heme and B) solid tumor cell lines show sub-nanomolar growth inhibition (CTG assay, 96h) and induction of apoptosis (caspase 3/7 activity, 48h) in response to KT-253. AML, ALL and solid tumor indications that include small round blue cell tumor (**SRBCT**) types were among the most sensitive. These data demonstrate the potential for KT-253 development across a wide variety of heme and solid tumor indications. See Poster PB032 which demonstrates that KT-253 shows robust antitumor activity in Merkel Cell Carcinoma, a neuroendocrine SRBCT.

Figure 2: 38% of Pediatric Solid Tumors Show Complete and Durable Responses to KT-253

A)

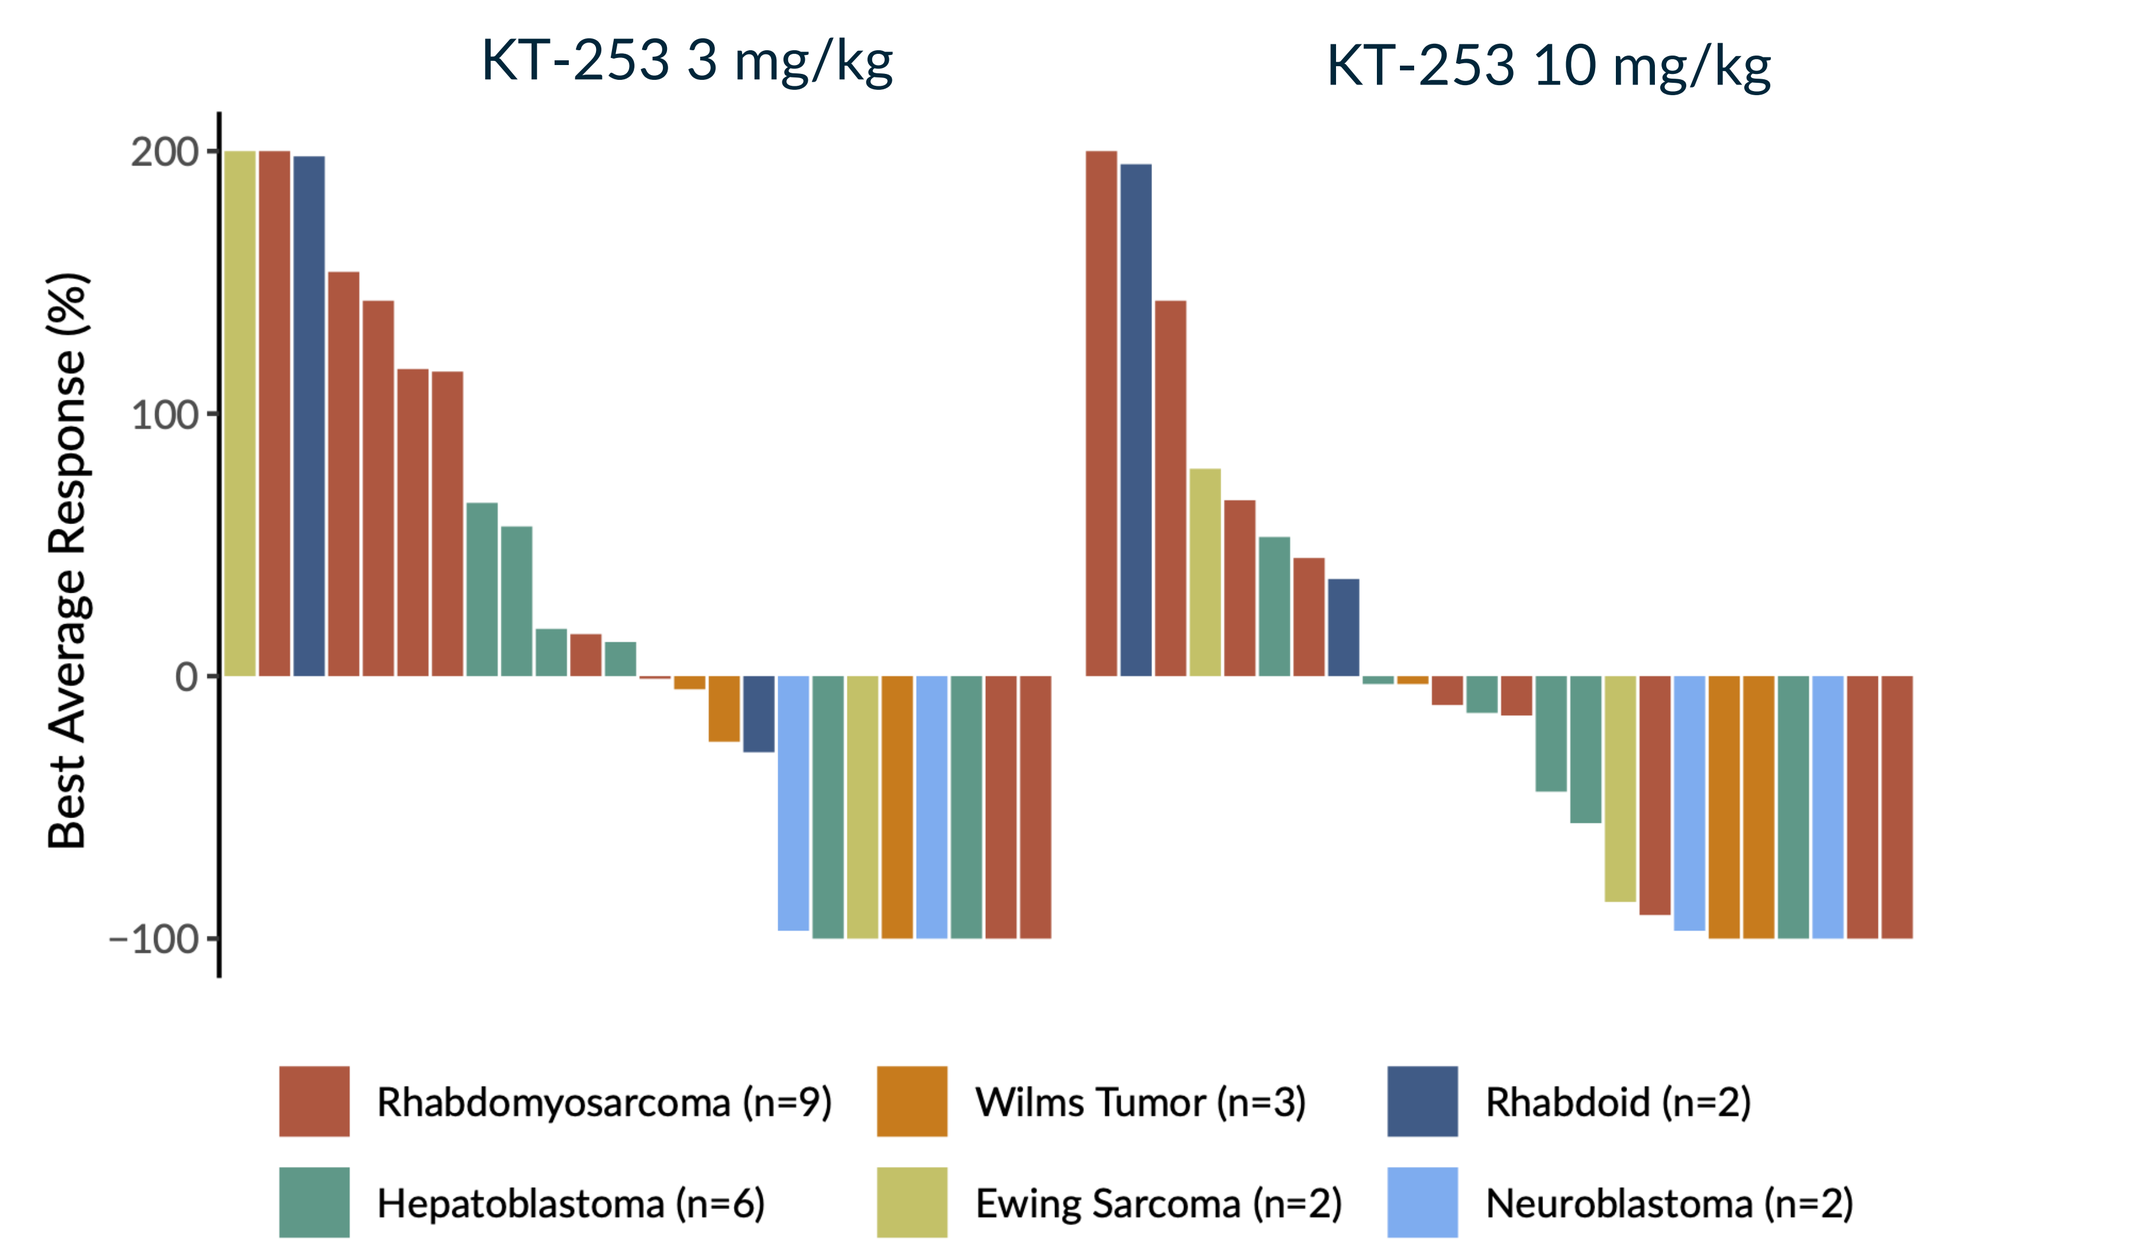
KT-253 3 mg/kg
KT-253 10 mg/kg
Best Average Response (%)
Rhabdomyosarcoma (n=9)
Wilms Tumor (n=3)
Rhabdoid (n=2)
Hepatoblastoma (n=6)
Ewing Sarcoma (n=2)
Neuroblastoma (n=2)

B)

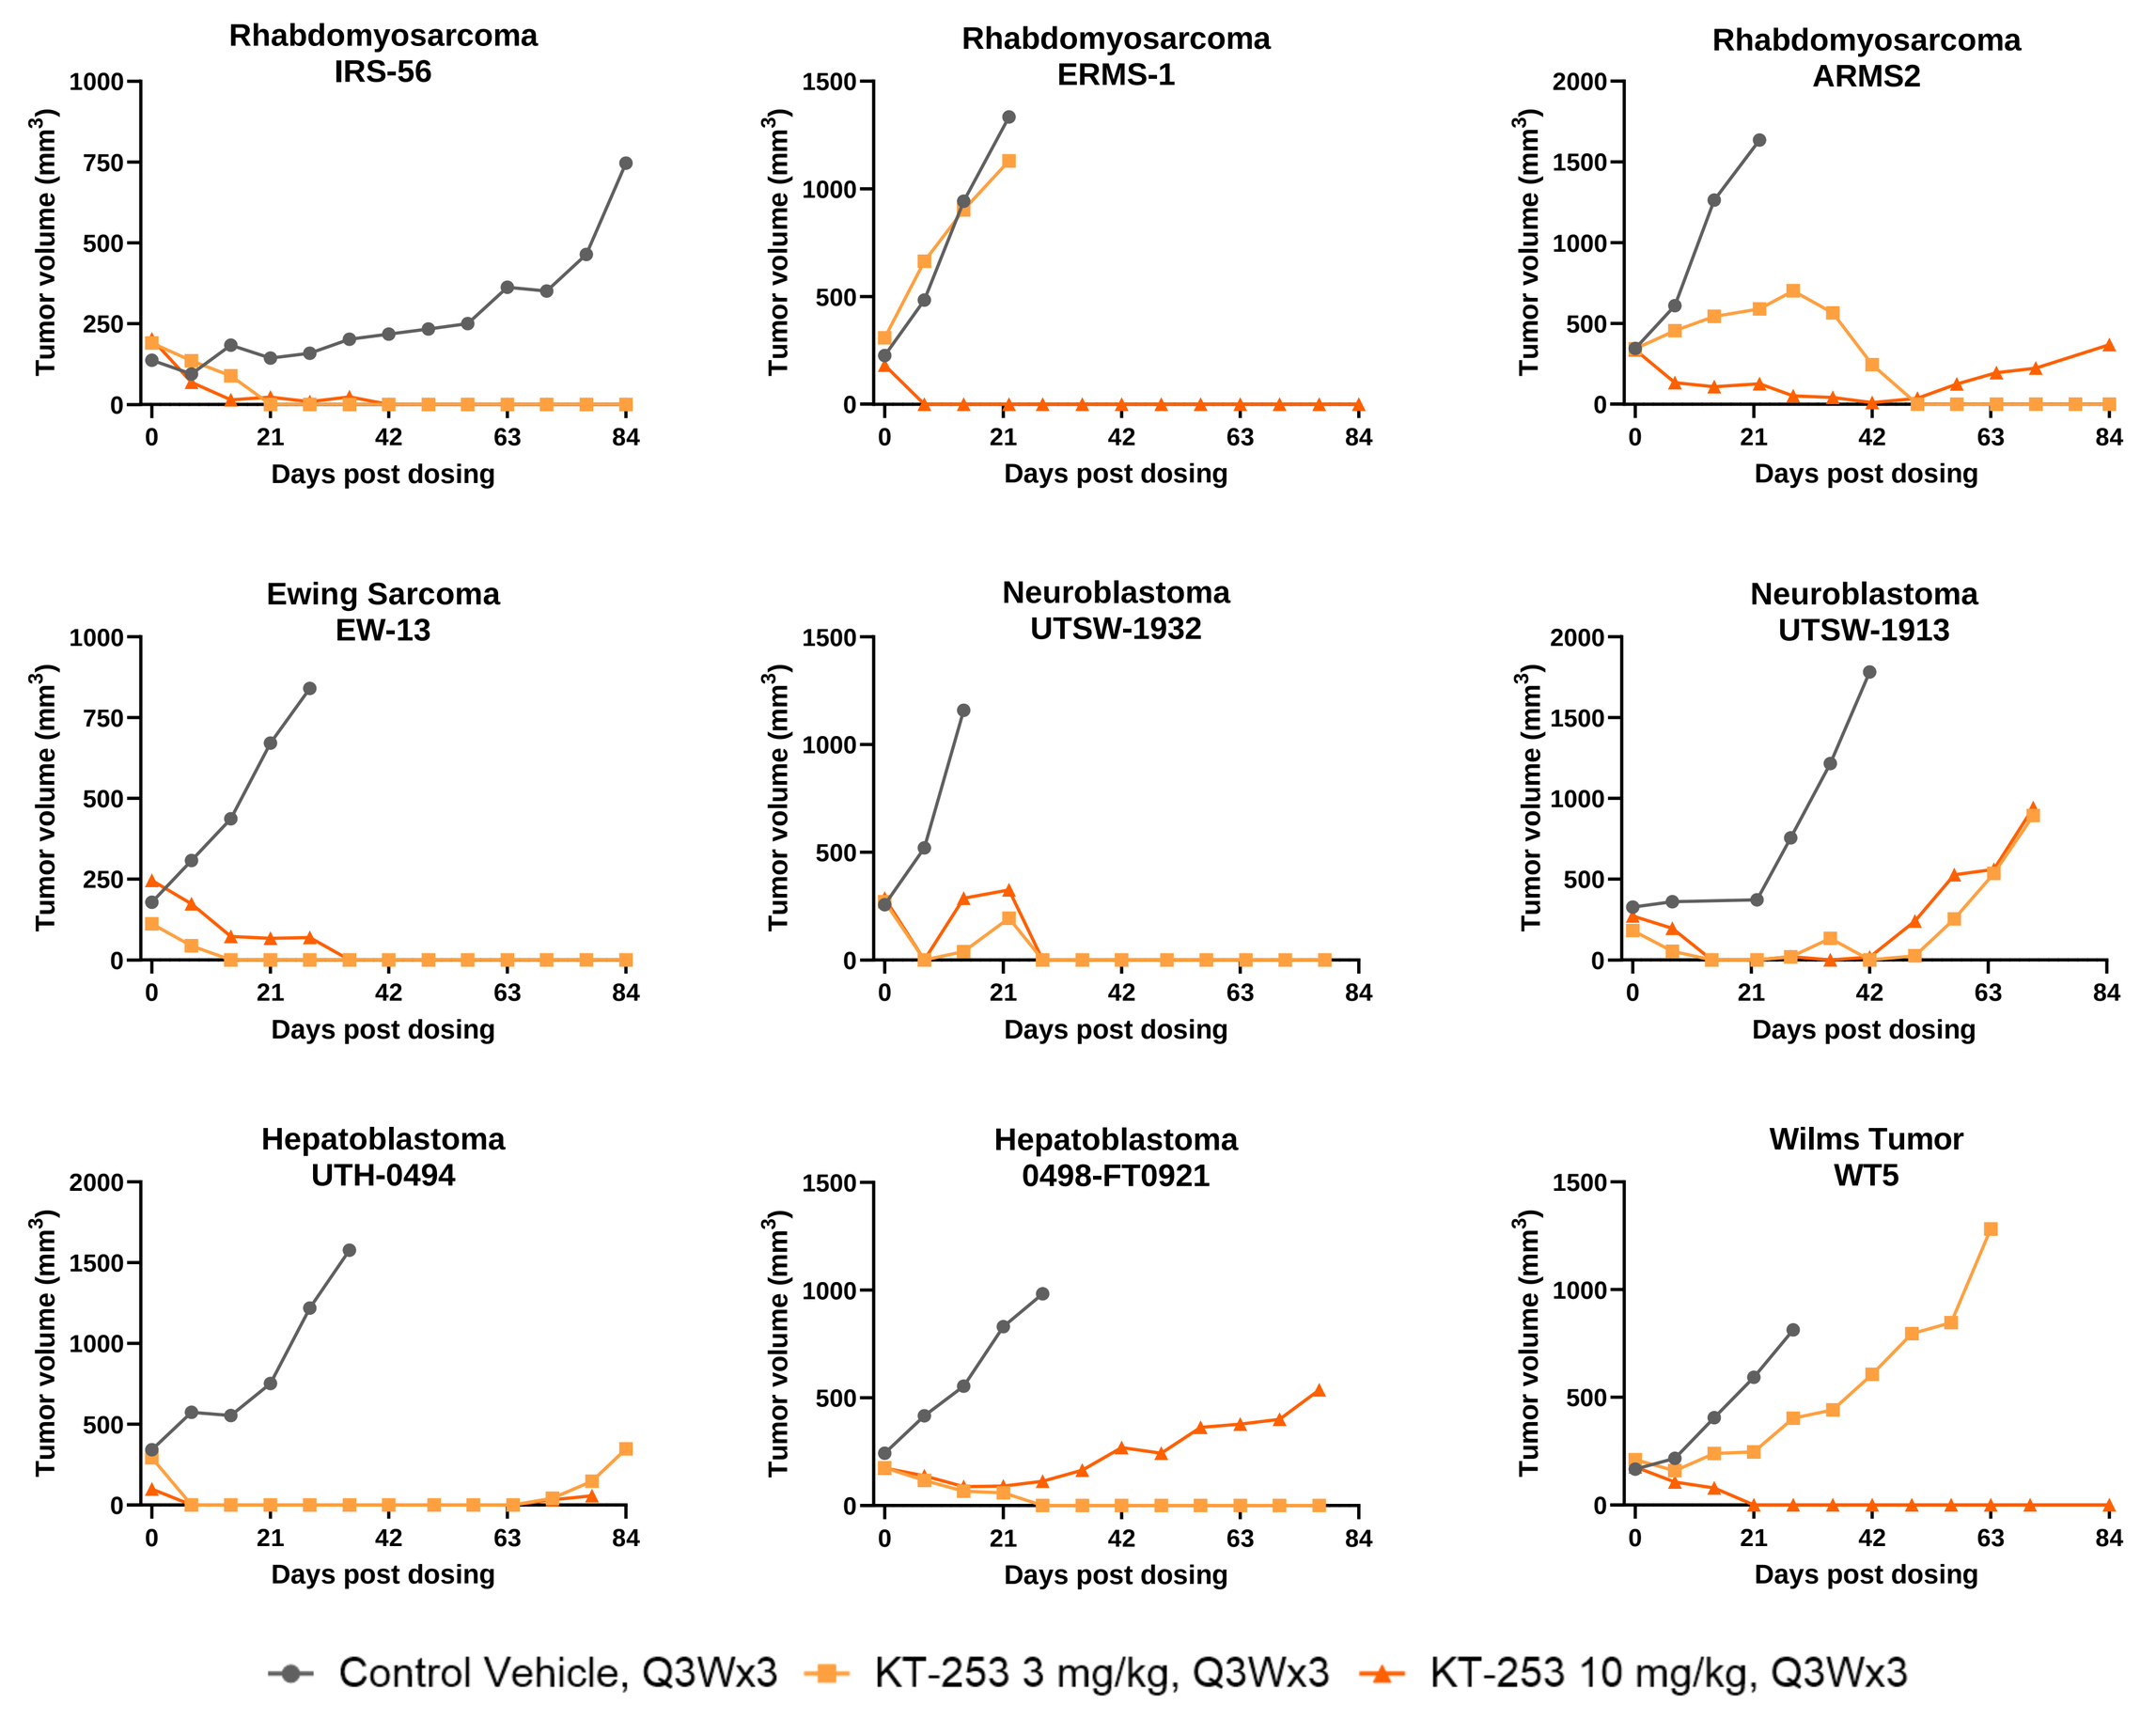
Rhabdomyosarcoma IRS-56
Rhabdomyosarcoma ERMS-1
Rhabdomyosarcoma ARMS2
Ewing Sarcoma EW-13
Neuroblastoma UTSW-1932
Neuroblastoma UTSW-1913
Hepatoblastoma UTH-0494
Hepatoblastoma 0498-FT0921
Wilms Tumor WT5
Tumor volume (mm3)
Days post dosing
Control Vehicle, Q3Wx3
KT-253 3 mg/kg, Q3Wx3
KT-253 10 mg/kg, Q3Wx3

A) Waterfall plot of best average response to KT-253 relative to baseline tumor volume at start of treatment in a single mouse trial of 24 pediatric PDX models representing 6 tumor types that include neuroendocrine and SRBCT types. Tumors were established subcutaneously in the hind flank of SCID mice. Complete responses were achieved in 9 of 24 models. Best average response is defined as the average of the best percent change in tumor volume over the course of 5 weeks.
B) Tumor growth curves for 9 models that showed complete response to KT-253 (n=1/group). Mice were dosed intravenously on day 0, 21, and 42.

Figure 3: KT-253 is Active in Multiple Adult Solid Tumor Types including Tumors that are Insensitive to an MDM2 Small Molecule Inhibitor

A)

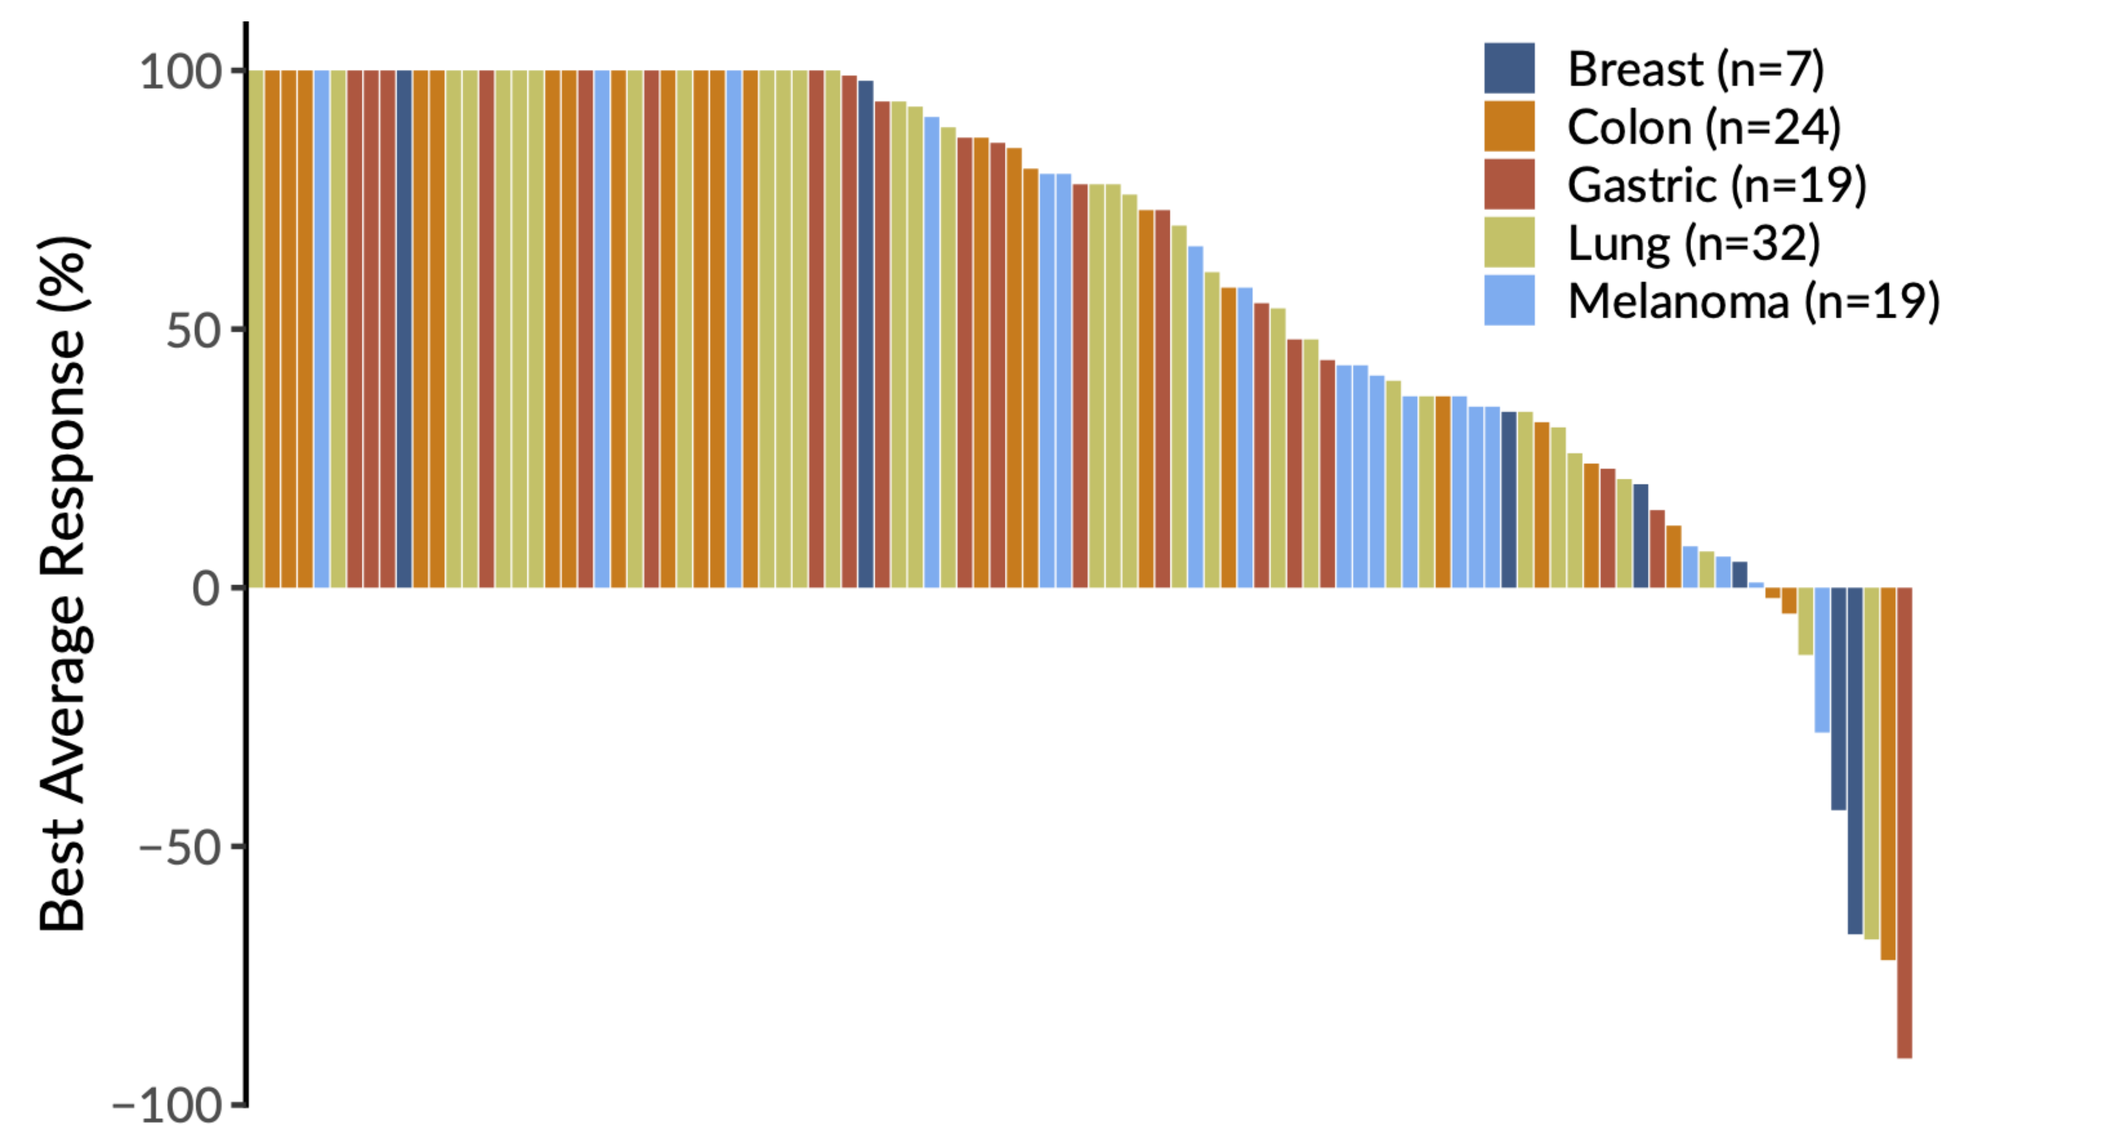
Best Average Response (%)
Breast (n=7)
Colon (n=24)
Gastric (n=19)
Lung (n=32)
Melanoma (n=19)

B)

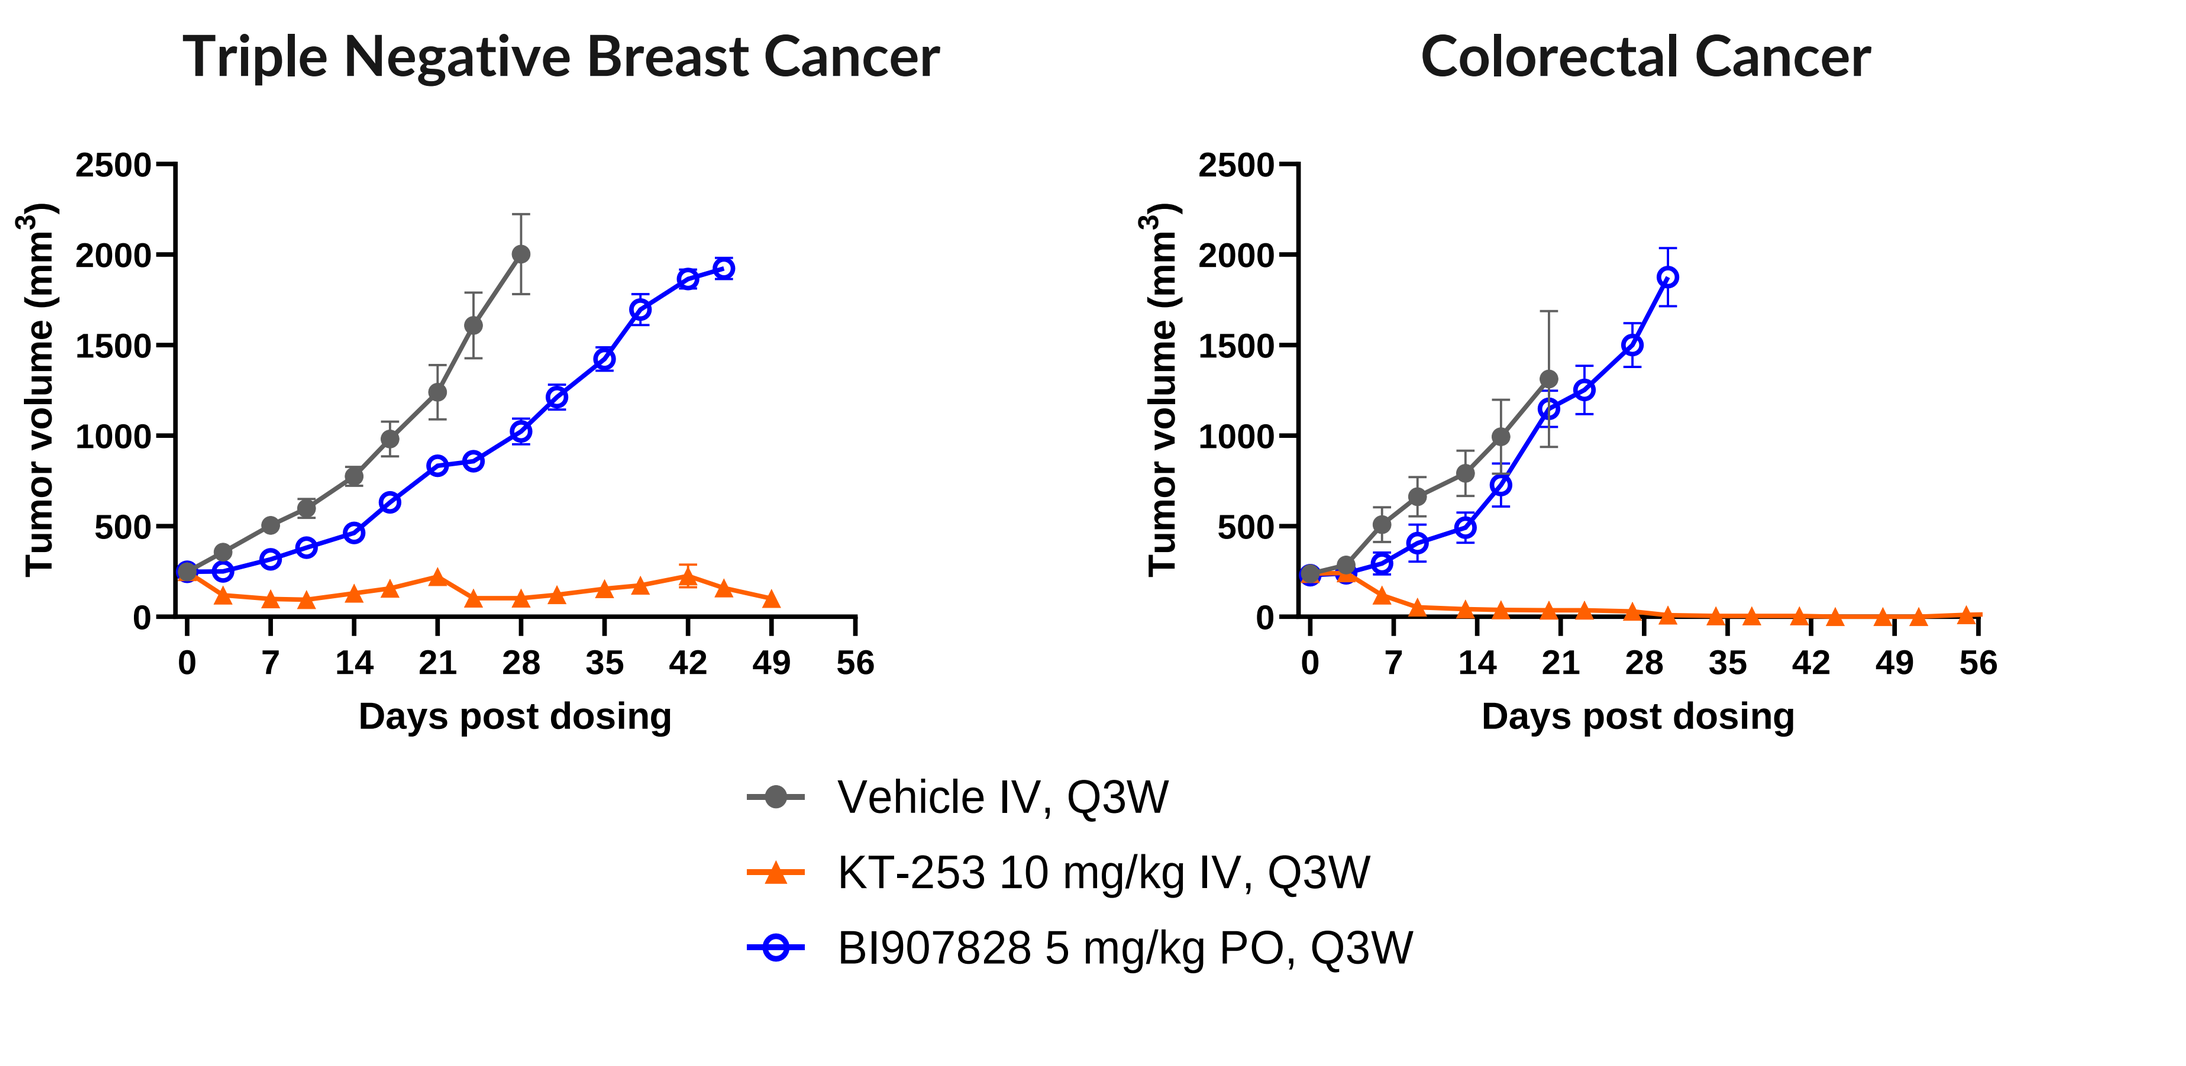
Triple Negative Breast Cancer
Colorectal Cancer
Tumor volume (mm3)
Days post dosing
Vehicle IV, Q3W
KT-253 10 mg/kg IV, Q3W
BI907828 5 mg/kg PO, Q3W

A) Waterfall plot of best average response to KT-253 relative to baseline tumor volume at start of treatment in a single mouse trial of 101 adult PDX models representing 5 different tumor types. Responders were observed in each tumor type. Best average response is defined as the average of the best percent change in tumor volume over the course of 5 tumor measurements.

B) Tumor growth curves comparing the activity of KT-253 to the MDM2 SMI, BI907828 in a triple negative breast cancer model and a colorectal cancer model that expresses the neuroendocrine marker synaptophysin. BI907828 was administered at the clinically relevant dose (n=3/group).

Figure 4: Finding a Gene Expression Signature that Predicts Response to KT-253

A)

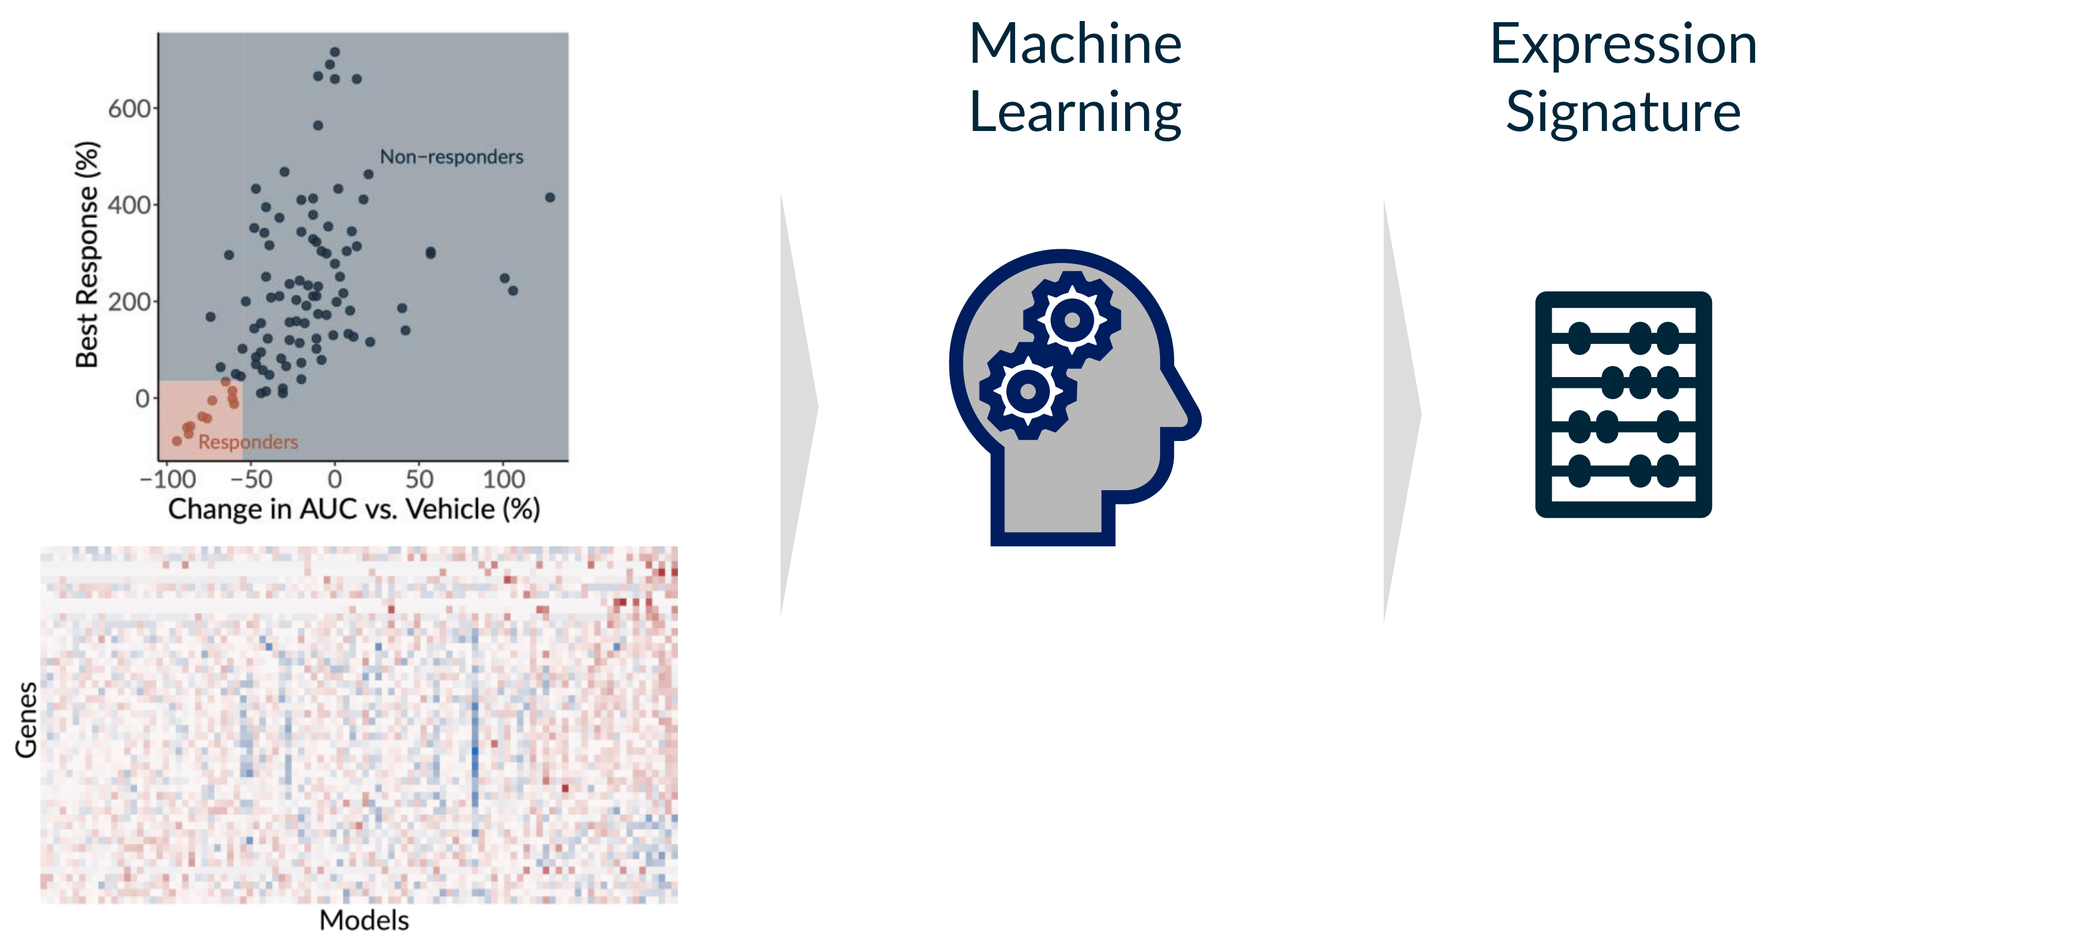
Best Response (%)
Non–responders
Responders
Change in AUC vs. Vehicle (%)
Genes
Models
Machine Learning
Expression Signature

B)

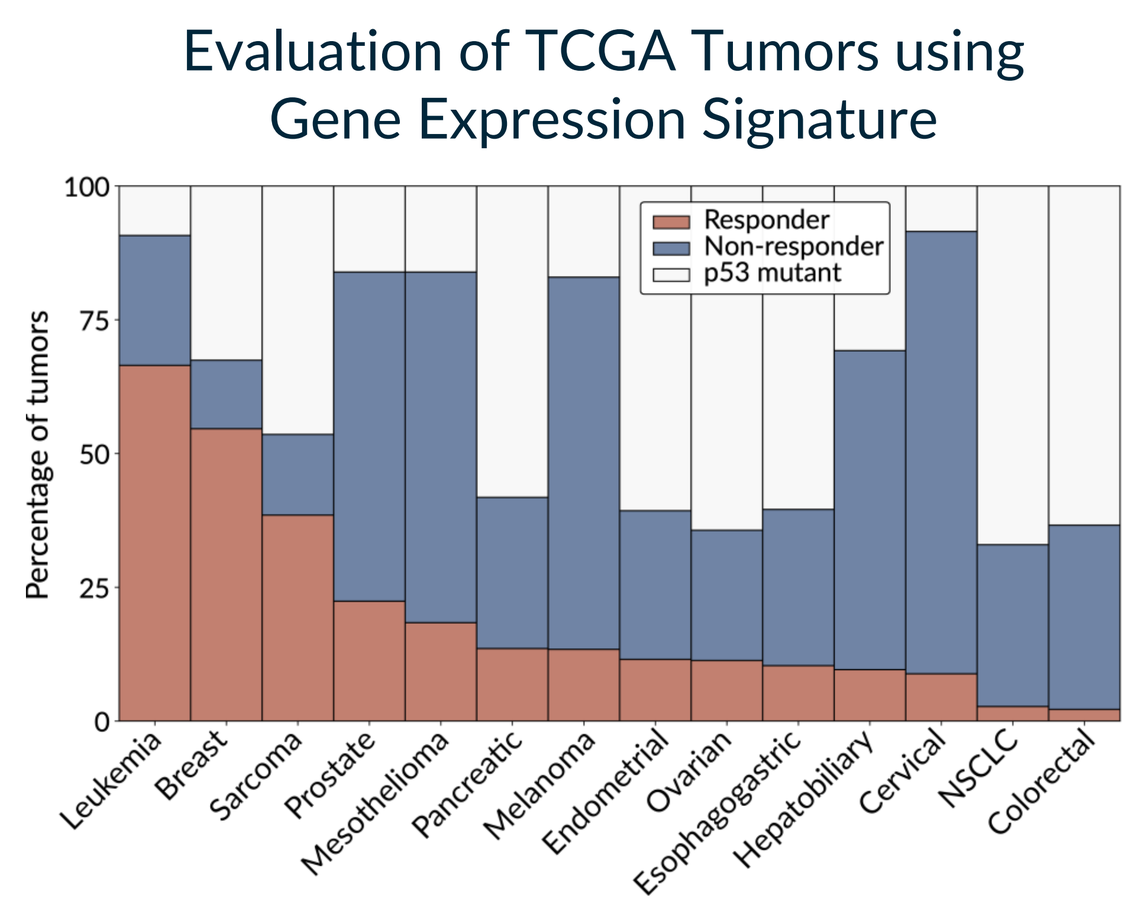
Evaluation of TCGA Tumors using Gene Expression Signature
Percentage of tumors
Responder
Non-responder
p53 mutant
Leukemia
Breast
Sarcoma
Prostate
Mesothelioma
Pancreatic
Melanoma
Endometrial
Ovarian
Esophagogastric
Hepatobiliary
Cervical
NSCLC
Colorectal

C)

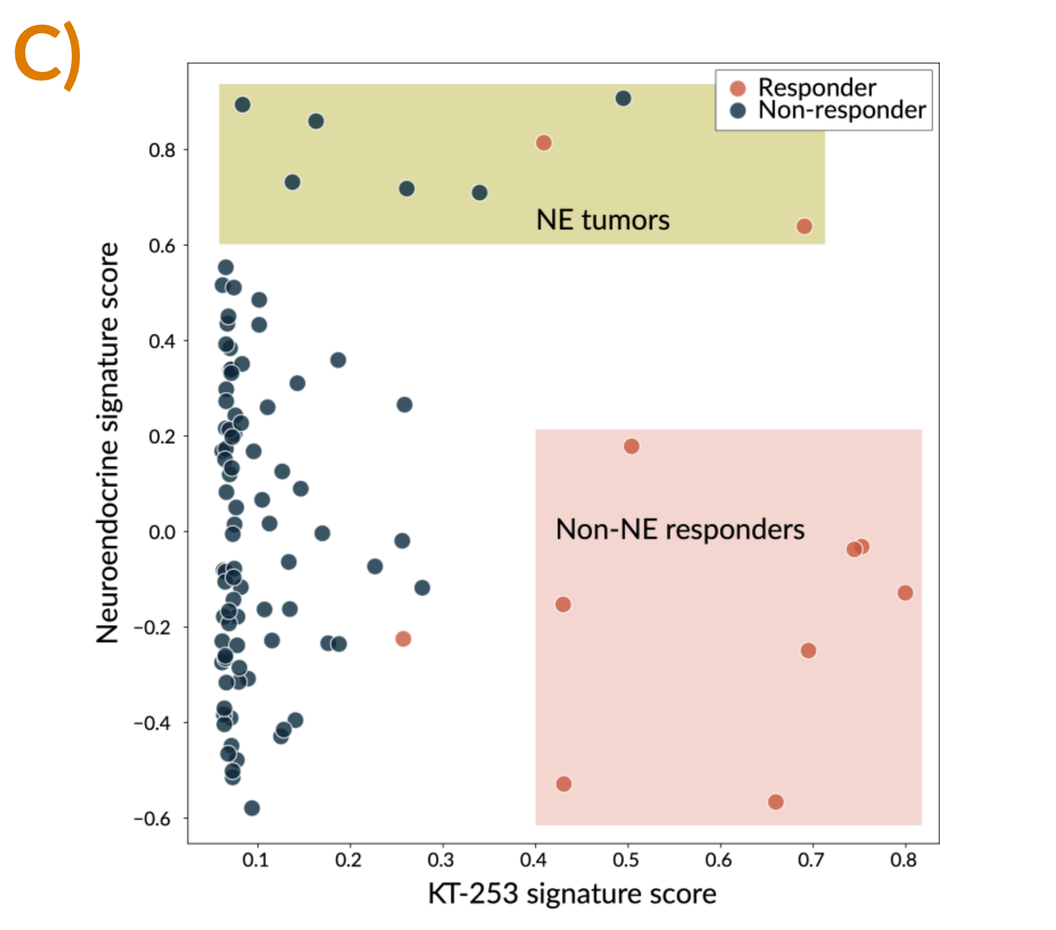
Responder
Non-responder
NE tumors
Non-NE responders
Neuroendocrine signature score
KT-253 signature score

A) To identify a broader pan-cancer responder signature, baseline expression profiles of the adult PDX models were used in a machine learning framework to build a predictive signature.

B) The signature was applied to The Cancer Genome Atlas (TCGA) pan-cancer dataset and identified AML and a subset of solid tumors as sensitive, consistent with our preclinical and early clinical findings.

C) Plot of a neuroendocrine (NE) signature (PMID: 29017058) vs. KT-253 predictive signature highlighting that the predictive signature identifies both neuroendocrine and non-endocrine tumors that respond to KT-253. The neuroendocrine tumors include 4 SCLC, 2 poorly differentiated gastric cancers, 1 melanoma and 1 synaptophysin positive CRC.

Figure 5: Disseminated AML PDX Models Show Complete Responses to KT-253

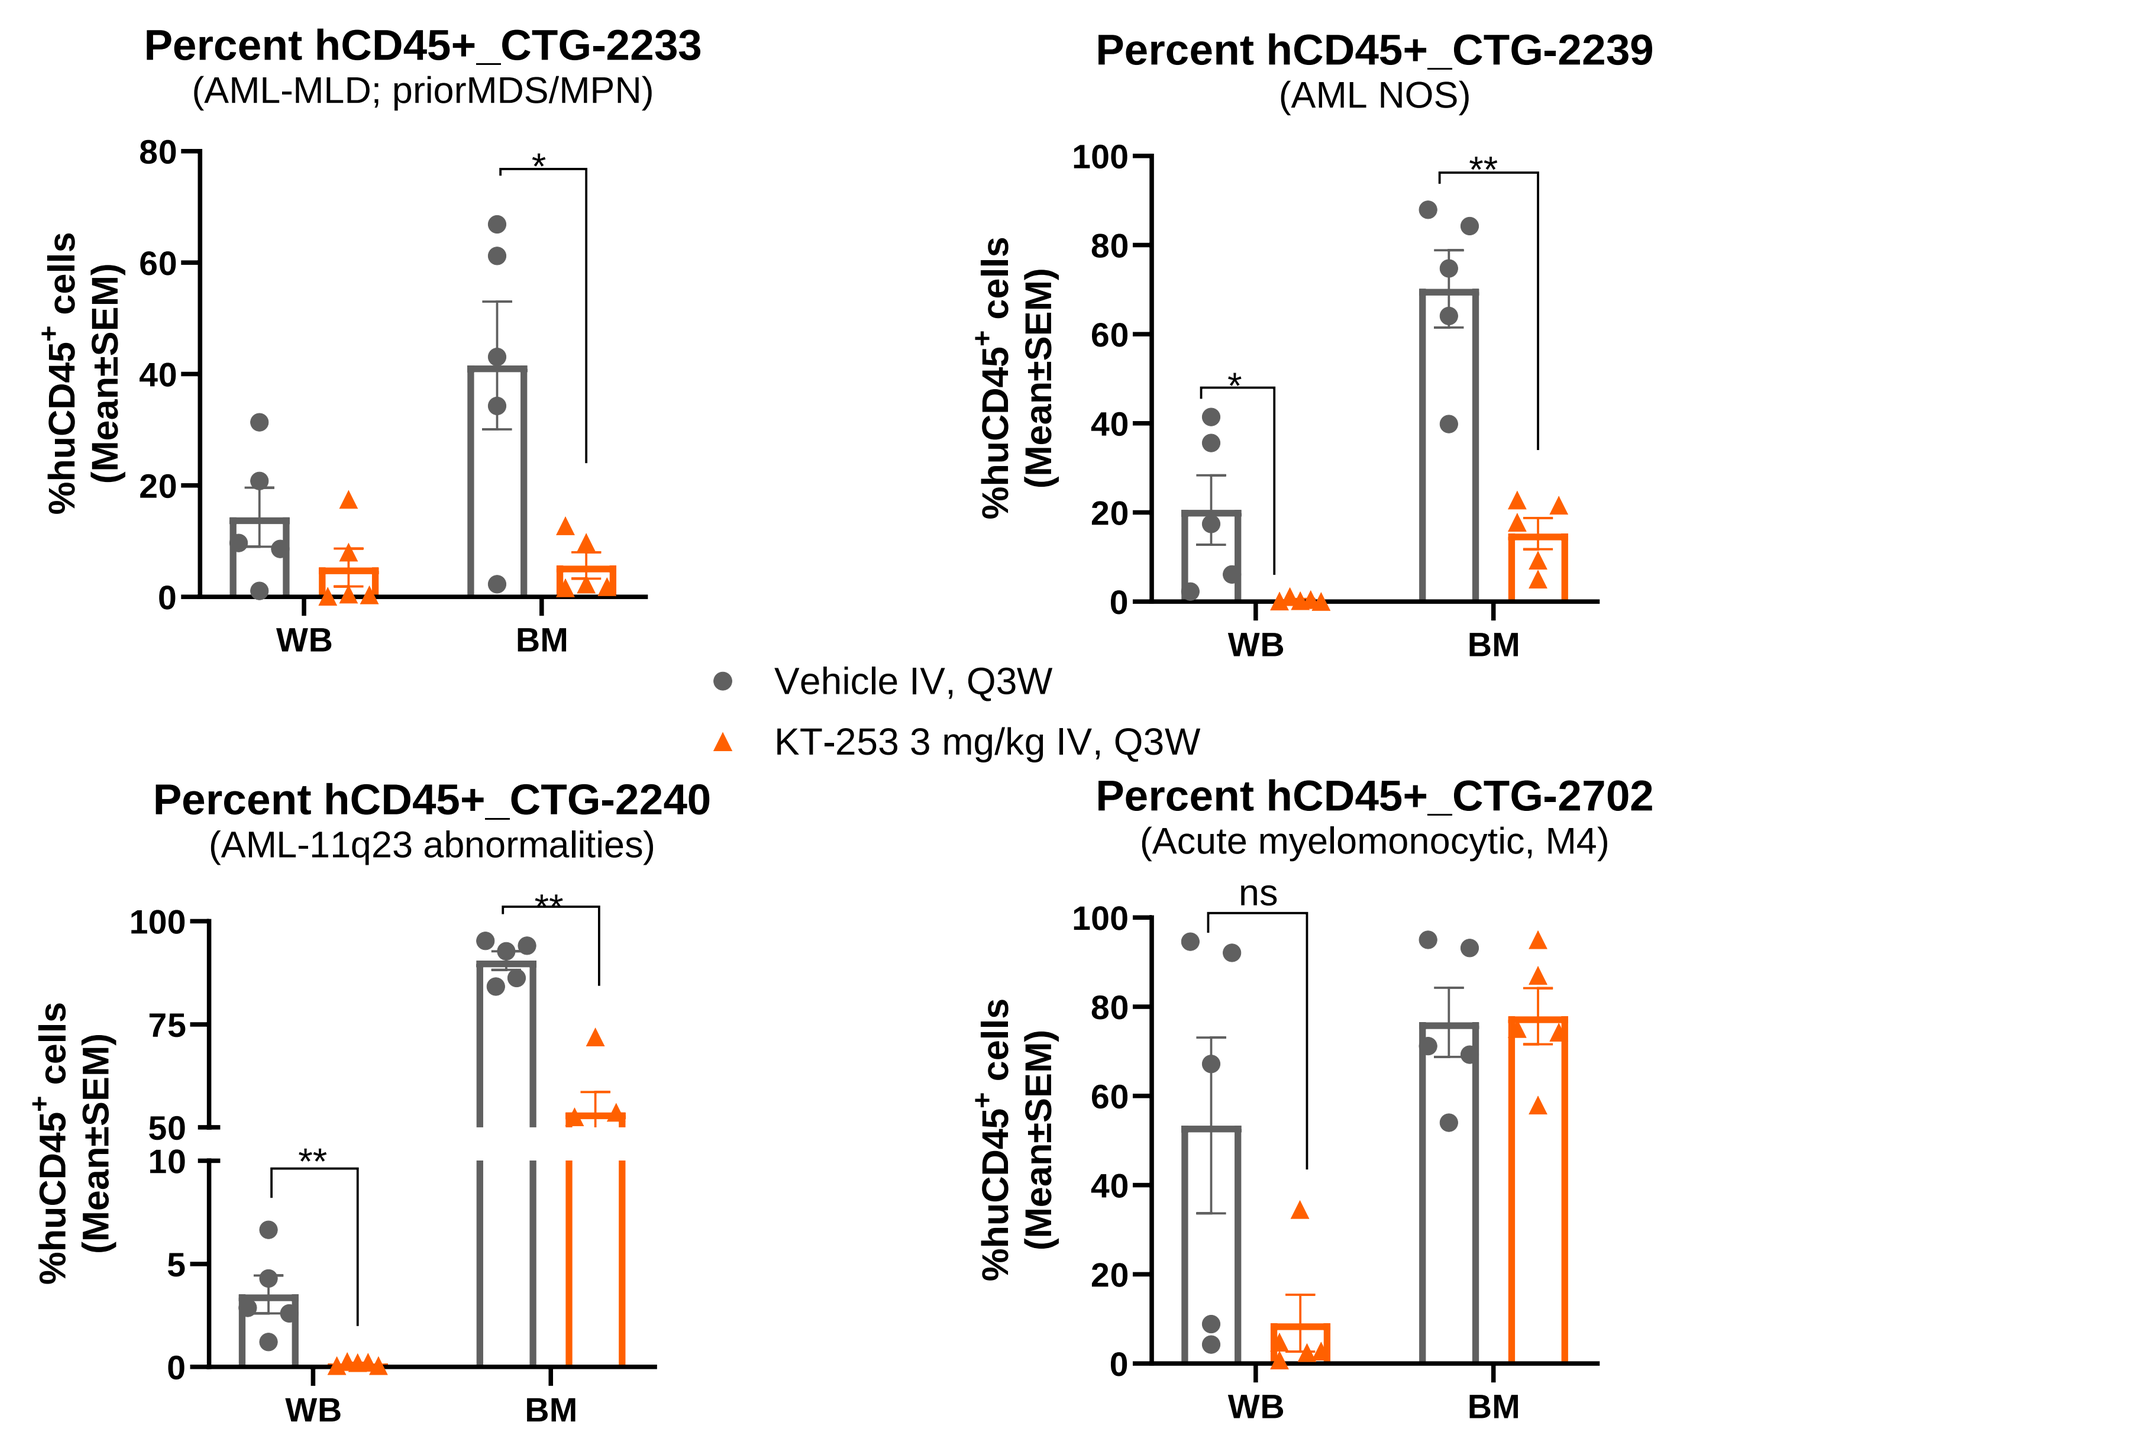
Percent hCD45+_CTG-2233
(AML-MLD; priorMDS/MPN)
Percent hCD45+_CTG-2239
(AML NOS)
Percent hCD45+_CTG-2240
(AML-11q23 abnormalities)
Percent hCD45+_CTG-2702
(Acute myelomonocytic, M4)
%huCD45+ cells (Mean±SEM)
WB
BM
Vehicle IV, Q3W
KT-253 3 mg/kg IV, Q3W

Bar graphs of the percent human CD45+ cells in whole blood (WB) and bone marrow (BM) after treatment with KT-253. Sub-lethally irradiated NOG-EXL mice were inoculated with 2x10^8 primary AML cells. Treatment was initiated when tumor engraftment levels in the bone marrow of surrogate animals were 68% (CTG-2233), 39% (CTG-2239), 21% (CTG-2240), 50% (CTG-2702). Mice were dosed on day 0, 21 and 42 and tumor burden was quantified by flow cytometry 24h after the last dose. Statistical analysis was conducted using a Student's t-test. Complete responses were achieved in 3 of the 4 models evaluated.

Figure 6: Prospectively Selected PDX Models Demonstrate Improved Response Rates and Survival

A)

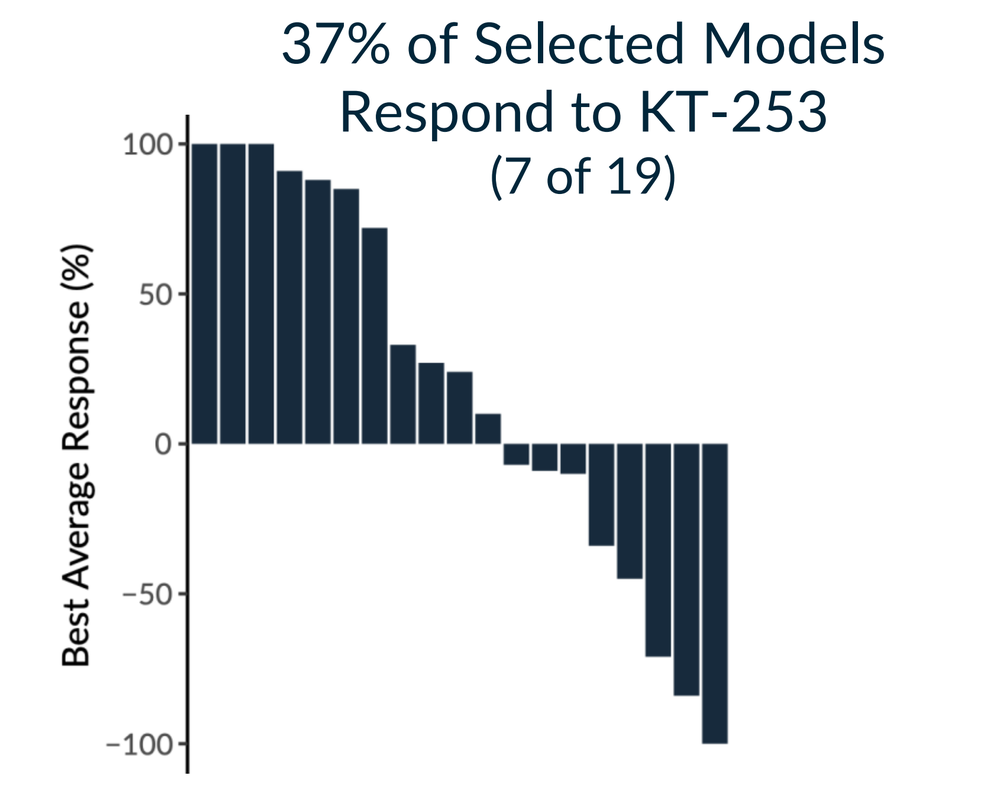
37% of Selected Models Respond to KT-253 (7 of 19)
Best Average Response (%)

B)

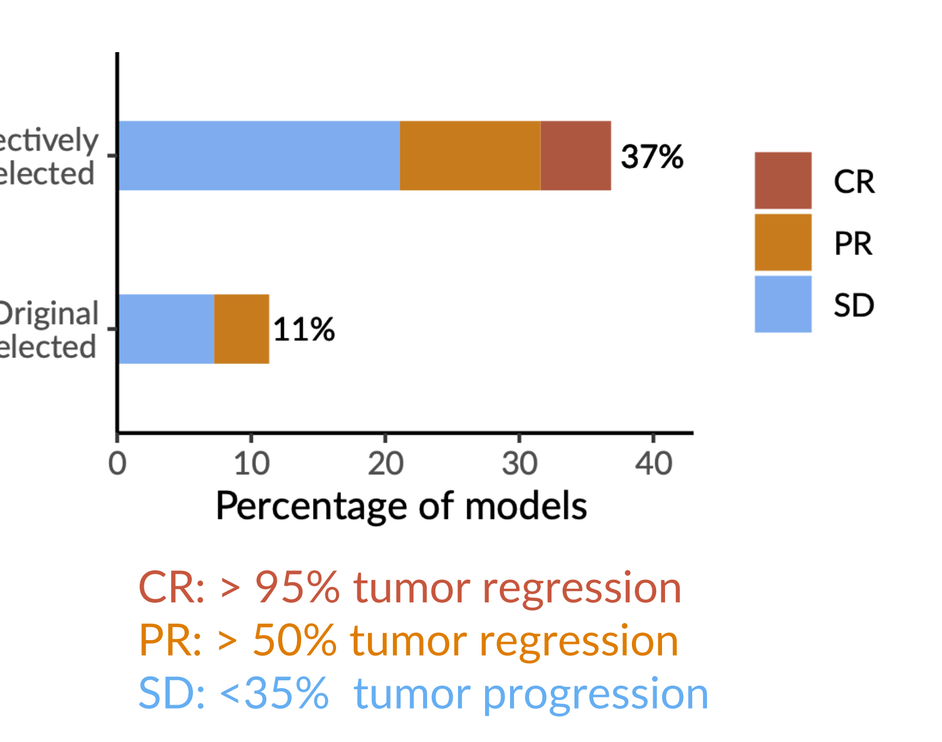
Prospectively Selected
Original Unselected
37%
11%
CR
PR
SD
Percentage of models
CR: > 95% tumor regression
PR: > 50% tumor regression
SD: <35% tumor progression

C)

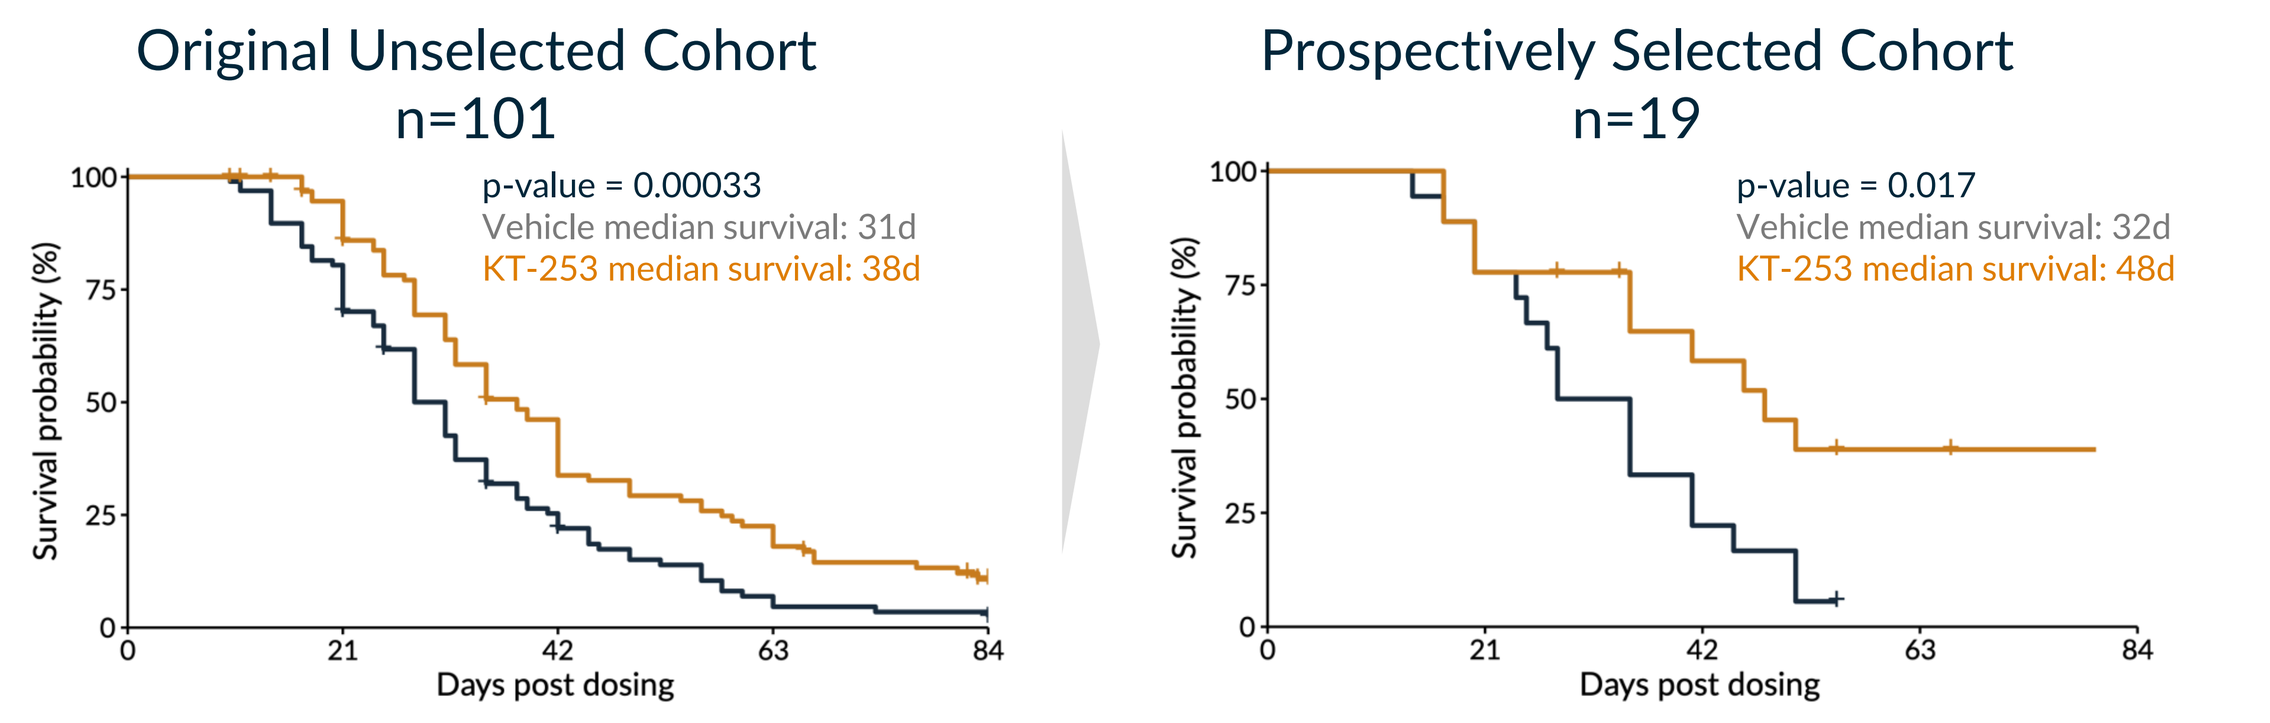
Original Unselected Cohort n=101
p-value = 0.00033
Vehicle median survival: 31d
KT-253 median survival: 38d
Prospectively Selected Cohort n=19
p-value = 0.017
Vehicle median survival: 32d
KT-253 median survival: 48d
Survival probability (%)
Days post dosing

A) Waterfall plot of best average response to KT-253 relative to baseline tumor volume at start of treatment in 19 adult PDX models selected using our predictive gene signature. 37% of prospectively selected models responded to KT-253 vs. 11% of unselected models.

B) Bar graph of response calls. Response is defined as the combination of 1) Best response after 14 days of SD or better, 2) Best average response over the course of 5 timepoints of SD or better, and 3) Less than -0.45 change in AUC relative to control. In unselected cohort, we observed 4 PR and 7 SD (11 of 101). In prospectively selected cohort, we observed 1 CR, 2 PR, 4 SD (7 of 19). CR: complete response, PR: partial response, SD: stable disease.

C) Kaplan-Meier curves comparing time to endpoint (tumor volume = 1500 mm^3) in control vs. KT-253 treated animals. Median survival was longer in the prospective models than in the original cohort (48d vs 38d). Odds ratio is 4.7 with 95% CI (1.29, 16.6). p-value= 0.0089.

CONCLUSIONS

- Single mouse trials show that KT-253 has the potential to induce durable complete responses in both pediatric and adult solid tumor types.
- We have used a machine learning framework to build a predictive signature of response to KT-253 that is distinct from published signatures.
- The resulting signature identifies AML and solid tumors with neuroendocrine characteristics as sensitive, consistent with our preclinical and early clinical findings. Importantly, it also identifies non-neuroendocrine tumors that respond to KT-253.
- Application of the responder signature for tumor model selection increased the KT-253 response rate in adult solid tumors from ~10 to 37% and prospectively selected models demonstrated prolonged median survival.
- The signature can be further refined using emerging clinical data to enrich for patients in future trials.

DISCLOSURES

This study was funded by Kymera Therapeutics. Dumont, Karnik, Chutake, Dey, Breitkopf, Howarth, Meeske, Fasciano, Mosher, Gollob and Williams are Kymera Therapeutics employees and equity owners.

The safety and efficacy of this investigational agent has not been established. This agent is not approved by any regulatory body for any indication.

KYMERA Kymera Therapeutics, 500 North Beacon Street, 4th Floor, Watertown, MA 02472 www.kymeratx.com Contact: Nancy Dumont / ndumont@kymeratx.com EORTC-NCI-AACR Symposium • October 23-25, 2024 • Barcelona, Spain