Safety, Pharmacokinetics, Pharmacodynamics and Clinical Activity of KT-333, a Targeted Protein Degrader of STAT3, in Patients with Relapsed or Refractory Hematologic and Solid Tumor Cancers

Aditi Shastri[1], Tatyana Feldman[2], Stefan K. Barta[3], Adam J. Olszewski[4], Auris O. Huen[5], Stephen D. Smith [6], Eric J. Feldman[1], Zachary D. Epstein-Peterson[7], Don A. Stevens[8], Pierluigi Porcu[9], Alexander N. Starodub[10], John C. Reneau[11], Jonathan E. Brammer[11], Ahmad Mattour[12], Lauren Pinter-Brown[13], Cristina P. Rodriguez[14], Ashwin Gollerkeri[16], Rachelle Perea[16], Joyoti Dey[16], Sean Donohue[16], Sagar Agarwal[16], Alyssa Fasciano[16], Jared Gollob[16], Enrica Marchi[17]

[1]Montefiore Medical Center and Albert Einstein College of Medicine, Bronx, NY, USA; [2]Hackensack University Medical Center John Theurer Cancer Center, Hackensack, NJ, USA; [3]Hospital of the University of Pennsylvania, Department of Medicine, Division of Hematology/Oncology, Philadelphia, PA, USA; [4]Lifespan Cancer Institute, Rhode Island; Hospital, Providence, RI, USA; [5]MD Anderson Cancer Center, Houston, Texas, USA; [6]Fred Hutchinson Cancer Center, Seattle, WA, USA; [1]Montefiore Medical Center and Albert Einstein College of Medicine, Bronx, NY, USA; [7]Memorial Sloan Kettering Cancer Center, New York, NY, USA; [8]Norton Cancer Institute, Louisville, KY, USA; [9]Thomas Jefferson University, Department of Medical Oncology, Philadelphia, PA, USA; [10]The Christ Hospital Cancer Center, Cincinnati, Ohio, USA; [11]The Ohio State University Wexner Medical Center, James Comprehensive Cancer Center, Columbus, OH, USA; [12]Henry Ford Cancer Institute, Detroit, MI, USA; [13]University of California Irvine, Chao Family Comprehensive Cancer Center, Orange, CA, USA; [14]University of Washington, Division of Medical Oncology, Seattle, WA, USA; [15]Kymera Therapeutics, Watertown, MA, USA; [16]University of Virginia Cancer Center, Program for T-Cell Lymphoma Research, Charlottesville, VA, USA

INTRODUCTION

STAT3

- STAT3 promotes tumor cell-intrinsic expression of genes involved with survival, proliferation, stemness and metastasis.
- STAT3 also promotes differentiation and activity of immunosuppressive cells in the tumor microenvironment.

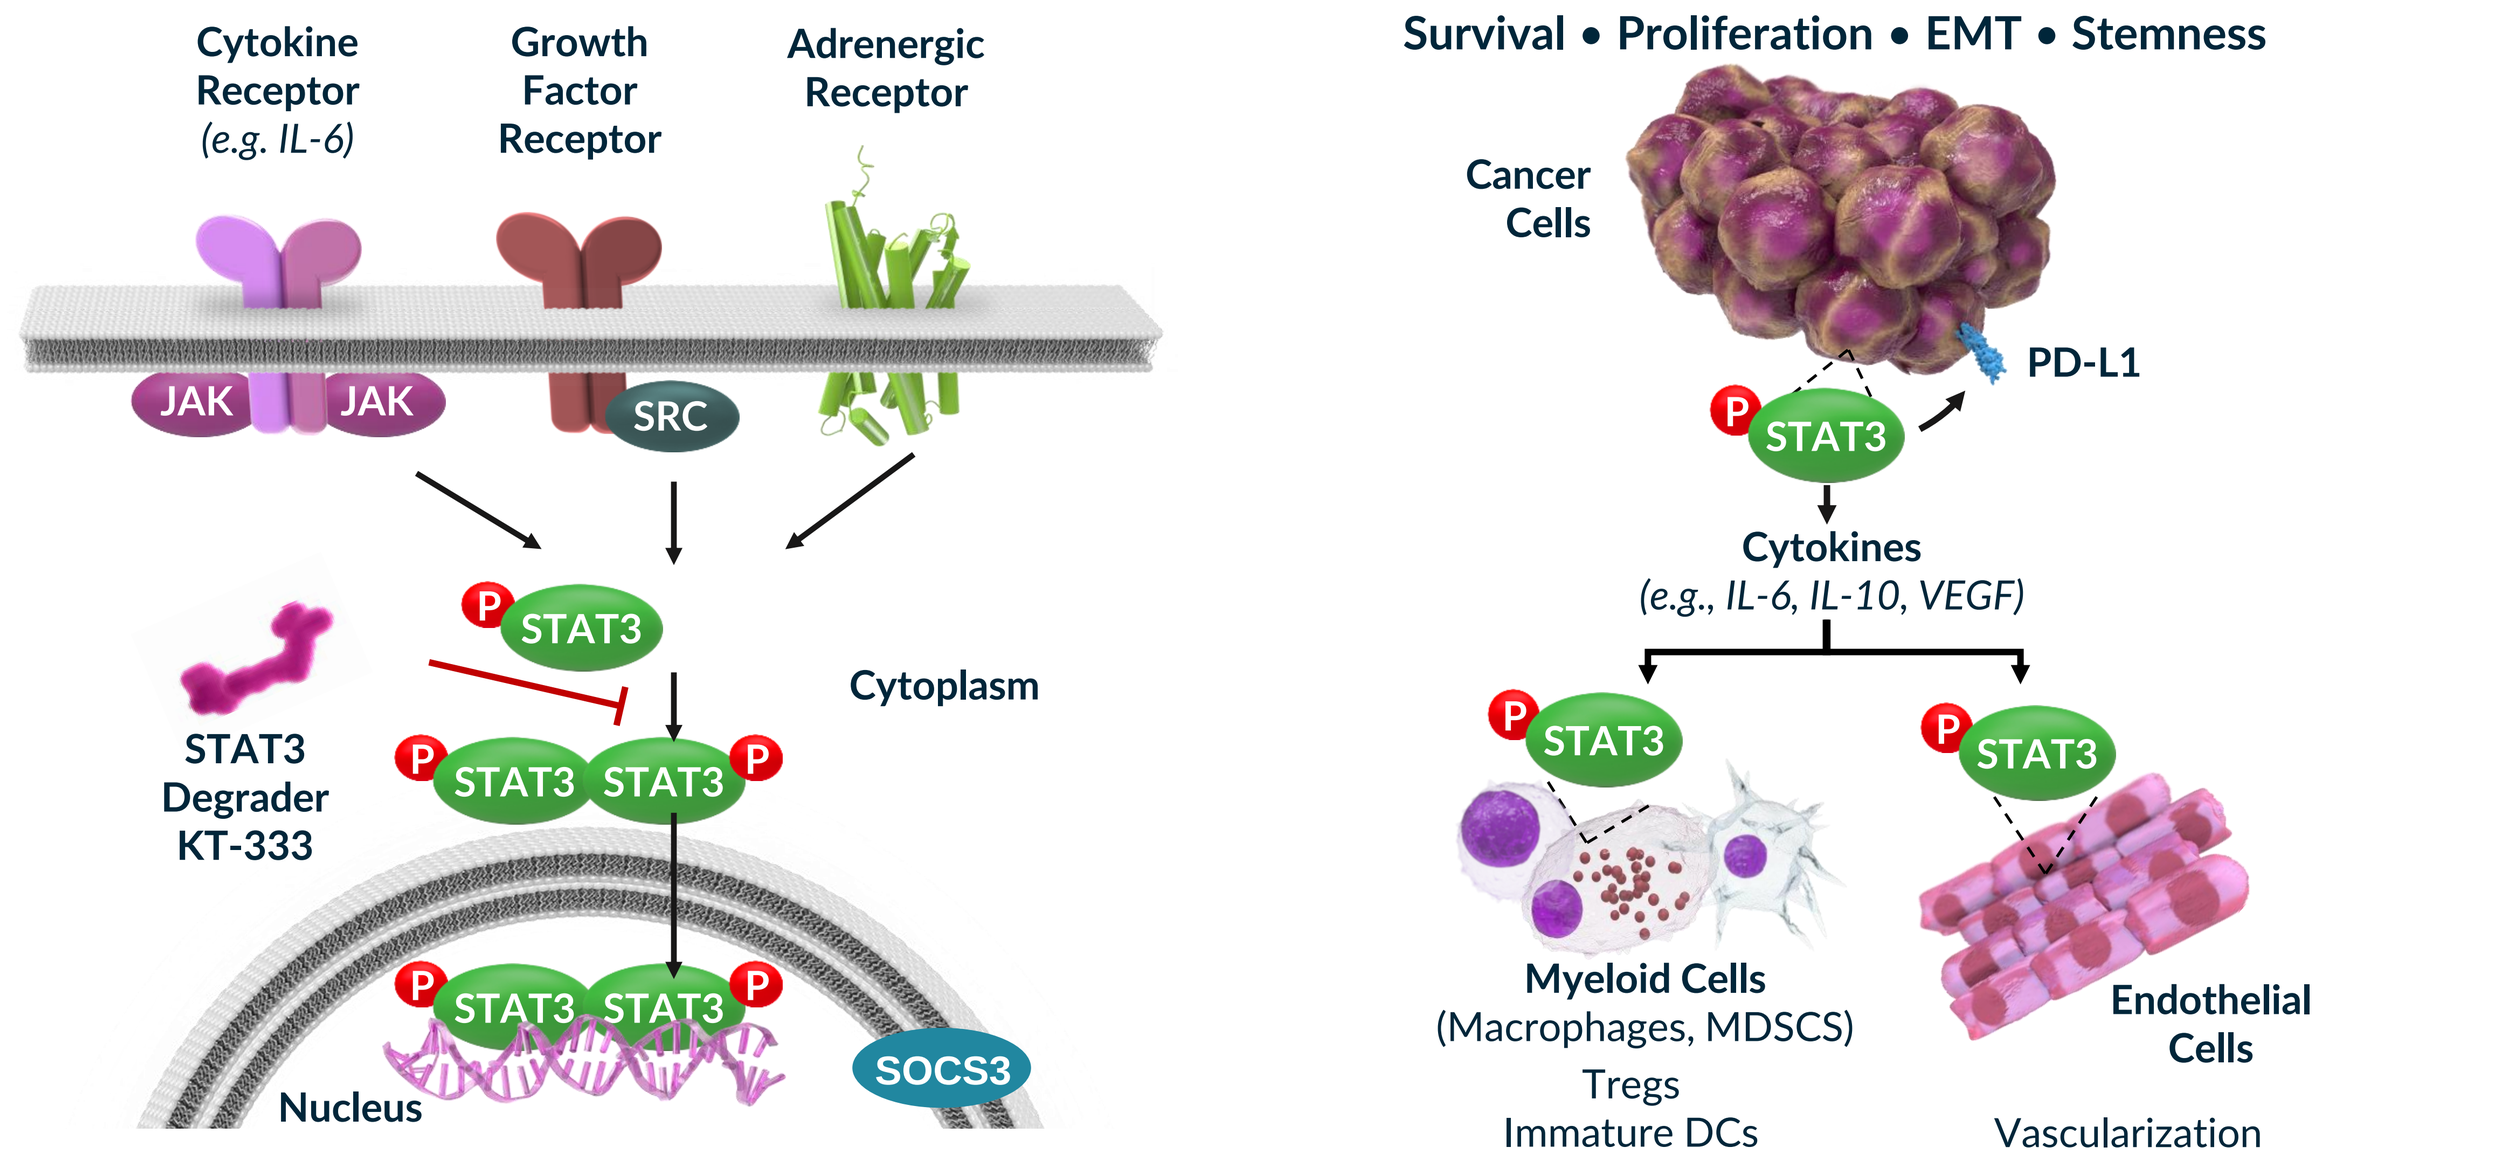
Cytokine Receptor (e.g. IL-6)
Growth Factor Receptor
Adrenergic Receptor
JAK
JAK
SRC
STAT3
STAT3 Degrader KT-333
STAT3 STAT3
Cytoplasm
STAT3 STAT3
SOCS3
Nucleus
Survival • Proliferation • EMT • Stemness
Cancer Cells
PD-L1
STAT3
Cytokines (e.g., IL-6, IL-10, VEGF)
STAT3
STAT3
Myeloid Cells (Macrophages, MDSCS)
Tregs
Immature DCs
Endothelial Cells
Vascularization

KT-333

- Targeted protein degraders are a new therapeutic class of compounds that utilize the ubiquitin proteasome system to target degradation of specific proteins.
- KT-333 is a first-in-class, potent, highly selective, heterobifunctional small molecule degrader of STAT3.
- In preclinical studies, proof of concept antitumor activity was seen with KT-333 monotherapy in mouse xenograft models of STAT3-dependent peripheral T-cell lymphoma (PTCL) and cutaneous T-cell lymphoma (CTCL). STAT3 degradation also led to an IFNγ response and TME remodeling in a syngeneic solid tumor model sensitizing to PD-1 blockade.

METHODS

Phase 1a Dose Escalation & MTD/RP2D Expansion

Phase 1b Dose Expansion Opportunities

KT-333 IV Weekly in 28-day Cycles

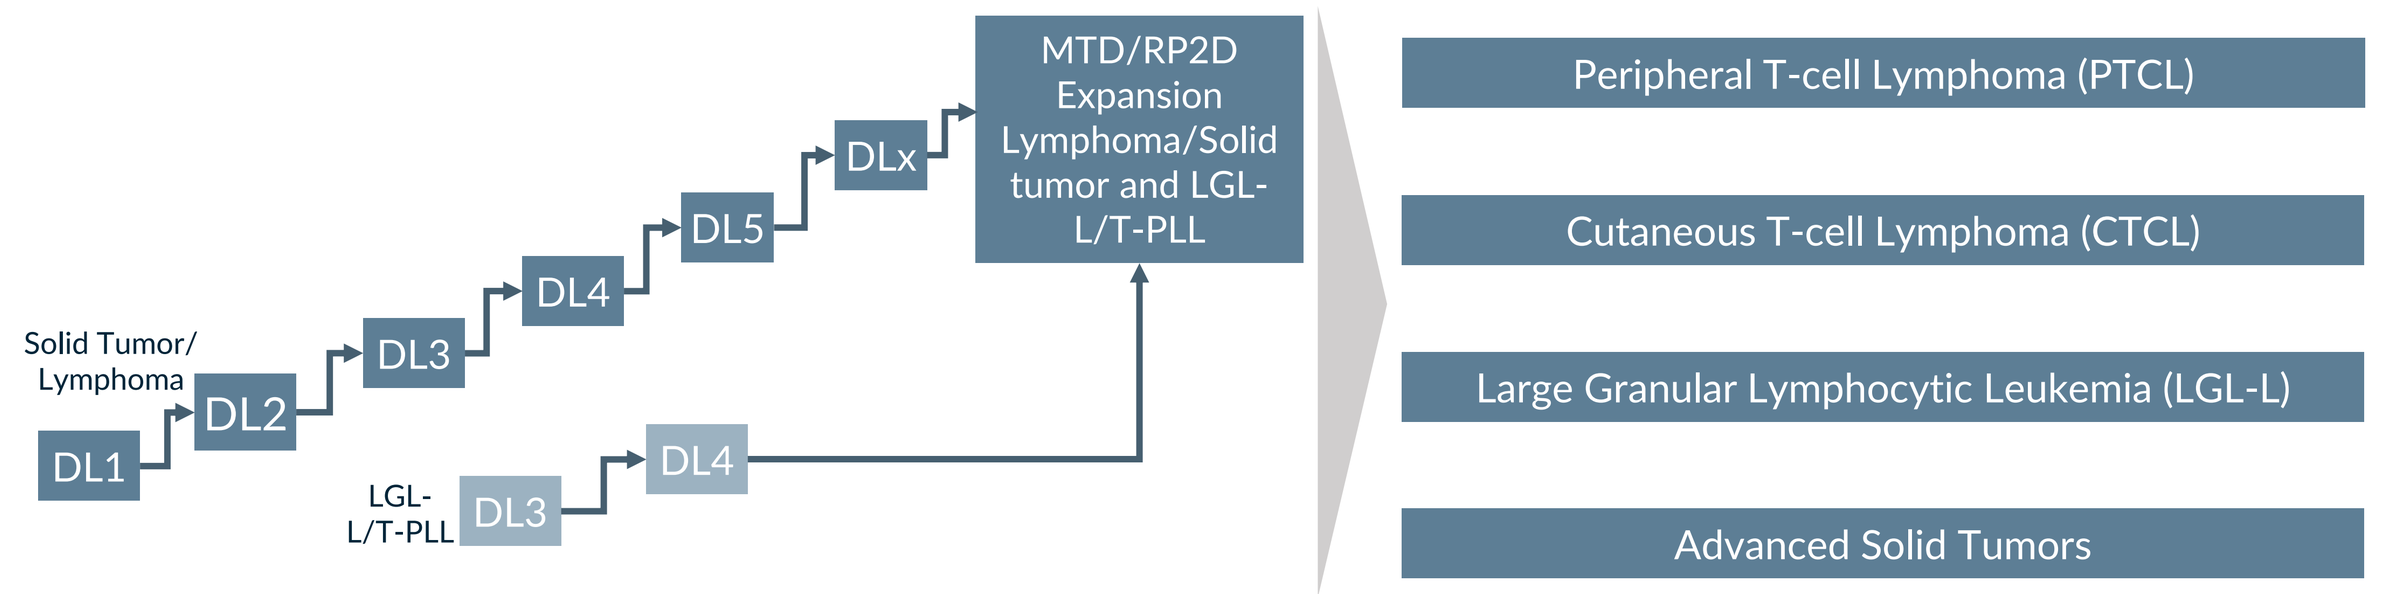
Solid Tumor/ Lymphoma
DL1
DL2
DL3
DL4
DL5
DLx
MTD/RP2D Expansion Lymphoma/Solid tumor and LGL-L/T-PLL
LGL-L/T-PLL
DL3
DL4
Peripheral T-cell Lymphoma (PTCL)
Cutaneous T-cell Lymphoma (CTCL)
Large Granular Lymphocytic Leukemia (LGL-L)
Advanced Solid Tumors

MTD=maximum tolerated dose; RP2D=recommended phase 2 dose; IV=intravenous; DL=dose level; T-PLL=T-cell prolymphocytic leukemia

Study Design and Objectives

Primary Objective:

- **Phase 1a.** Overall safety profile of escalating doses of KT-333 and determination of the maximum tolerated dose (MTD)/recommended Phase 2 dose (RP2D).
- **Phase 1b.** Safety and tolerability of KT-333 at the RP2D in patients with PTCL, LGL-L, CTCL and solid tumors.

Secondary Objective: PK and preliminary clinical activity.

Exploratory: STAT3 degradation and STAT3-regulated circulating biomarkers in peripheral blood; STAT3/pSTAT3 expression and immune TME profiling in baseline and on-treatment tumor biopsies; Gene expression in peripheral blood and tumor biopsy; STAT3 mutational analyses.

Key Eligibility Criteria

Inclusion Criteria:

- **Phase 1a.**
 - Lymphomas (including Hodgkin, B-, T- and NK-Cell lymphoma) or solid tumors relapsed/refractory (R/R) to at least two prior treatments or with no available standard therapy.
 - LGL-L/T-PLL: R/R to one prior systemic treatment.
- **Phase 1b.**
 - PTCL, CTCL, LGL-L (T-cell LGL-L or CLPD-NK) or solid tumors R/R to at least one prior systemic treatment or with no available standard therapy.
- ECOG of 0-2.
- Adequate liver/kidney (all patients) and bone marrow function (except for LGL-L).

Exclusion Criteria:

- Radiation, anti-cancer therapy or major surgery within 4 weeks.
- Autologous hematopoietic stem cell transplant less than 3 months prior to first dose of study drug.
- Allogenic hematopoietic or bone marrow transplant less than 6 months prior to 1st dose.
- Diagnosis of Chronic Lymphocytic Leukemia or small lymphocytic leukemia.

RESULTS

Demographics

	Dose Level 1 0.05 mg/kg (n=4)	Dose Level 2 0.1 mg/kg (n=4)	Dose Level 3 0.2 mg/kg (n=8)	Dose Level 4 0.4 mg/kg (n=14)	Dose Level 5 0.7 mg/kg (n=8)	Dose Level 6 1.1 mg/kg (n=6)	Dose Level 7 1.5 mg/kg (n=3)	Overall (N=47)
Age (years)								
Median (min, max)	64.5 (57, 70)	63.5 (59, 74)	70.5 (40, 76)	63.5 (42, 81)	66.0 (30, 75)	45.5 (24, 73)	61.0 (50, 65)	65.0 (24, 81)
Sex (n, (%))								
Male	3 (75.0)	1 (25.0)	4 (50.0)	12 (85.7)	6 (75.0)	2 (33.3)	-	28 (59.6)
ECOG								
0	1 (25.0)	-	4 (50.0)	5 (35.7)	4 (50.0)	4 (66.7)	1 (33.3)	19 (40.4)
1	3 (75.0)	4 (100)	4 (50.0)	9 (64.3)	4 (50.0)	1 (16.7)	2 (66.7)	27 (57.4)
2	-	-	-	-	-	1 (16.7)	-	1 (2.1)
Prior Systemic Therapy Regimens								
≥4	2 (50.0)	4 (100.0)	5 (62.5)	7 (50.0)	3 (37.5)	4 (66.7)	2 (66.7)	26 (55.3)
Tumor Type								
Solid Tumor‡	3 (75.0)	2 (50.0)	5 (62.5)	7 (50.0)	3 (37.5)	-	1 (33.3)	21 (44.7)
CTCL	1 (25.0)	1 (25.0)	-	3 (21.4)	2 (25.0)	4 (66.7)	-	11 (23.4)
T-Cell LGL-L	-	-	2 (25.0)	-	2 (25.0)	-	-	4 (8.5)
Hodgkin's	-	-	-	2 (14.3)	-	2 (33.3)	-	4 (8.5)
PTCL	-	1 (25.0)	-	1 (7.1)	-	-	1 (33.3)	3 (6.4)
T-PLL	-	-	1 (12.5)	1 (7.1)	-	-	-	2 (4.3)
NK-Cell Lymphoma	-	-	-	-	-	-	1 (33.3)	1 (2.1)
B-Cell Lymphoma	-	-	-	-	1 (12.5)	-	-	1 (2.1)

‡ = colorectal (4); head and neck (3); pancreatic (2); anal; appendiceal; cervical; cholangiocarcinoma; colon adenocarcinoma; duodenal; endometrial; gallbladder; ovarian, peritoneal, rectal and renal (n=1 each)

Overall Safety

Number of Patients with Adverse Event Occurring in >10 % Patients (n, (%)) – Regardless of Causality

Preferred Term	Dose Level 1 0.05 mg/kg (n=4)		Dose Level 2 0.1 mg/kg (n=4)		Dose Level 3 0.2 mg/kg (n=8)		Dose Level 4 0.4 mg/kg (n=14)		Dose Level 5 0.7 mg/kg (n=8)		Dose Level 6 1.1 mg/kg (n=6)		Dose Level 7 1.5 mg/kg (n=3)		Overall (N=47)	
	All	≥Grade 3	All	≥Grade 3	All	≥Grade 3	All	≥Grade 3	All	≥Grade 3	All	≥Grade 3	All	≥Grade 3	All	≥Grade 3*
Stomatitis	-	-	-	-	3 (37.5)	-	4 (28.6)	-	6 (75.0)	1 (12.5)	5 (83.3)	1 (16.7)	2 (66.7)	-	20 (42.6)	2 (4.3)
Fatigue	2 (50.0)	1 (25.0)	1 (25.0)	-	3 (37.5)	-	2 (14.3)	-	-	-	2 (33.3)	-	2 (66.7)	1 (33.3)	12 (25.5)	2 (4.3)
Nausea	1 (25.0)	-	1 (25.0)	-	3 (37.5)	-	3 (21.4)	-	2 (25.0)	-	1 (16.7)	-	1 (33.3)	-	12 (25.5)	-
Pyrexia	-	-	-	-	-	-	2 (14.3)	-	3 (37.5)	-	2 (33.3)	-	2 (66.7)	-	9 (19.1)	-
ALT increased	-	-	-	-	2 (25.0)	1 (12.5)	4 (28.6)	-	2 (25.0)	-	-	-	-	-	8 (17.0)	1 (2.1)
Constipation	2 (50.0)	-	1 (25.0)	-	-	-	3 (21.4)	-	2 (25.0)	-	-	-	-	-	8 (17.0)	-
Diarrhea	-	-	-	-	4 (50.0)	-	2 (14.3)	-	1 (12.5)	-	1 (16.7)	-	-	-	8 (17.0)	-
Anaemia	-	-	2 (50.0)	-	1 (12.5)	-	2 (14.3)	2 (14.3)	1 (12.5)	-	1 (16.7)	-	-	-	7 (14.9)	2 (4.3)
AST increased	-	-	-	-	2 (25.0)	1 (12.5)	3 (21.4)	-	2 (25.0)	-		-	-	-	7 (14.9)	1 (2.1)
Oropharyngeal pain	-	-	-	-	-	-	3 (21.4)	-	-	-	1 (16.7)	-	2 (66.7)	-	6 (12.8)	-
Pruritus	-	-	-	-	-	-	3 (21.4)	1 (7.1)	-	-	1 (16.7)	-	2 (66.7)	-	6 (12.8)	1 (2.1)
Abdominal pain	2 (50.0)	1 (25.0)	-	-	-	-	1 (7.1)	1 (7.1)	2 (25.0)	1 (12.5)		-		-	5 (10.6)	3 (6.4)
Chills	-	-	-	-	-	-	-	-	-	-	3 (50.0)	-	2 (66.7)	-	5 (10.6)	-
Cough	1 (25.0)	-	-	-	-	-	4 (28.6)	-	-	-	-	-	-	-	5 (10.6)	-
Decreased appetite	-	-	1 (25.0)	-	2 (25.0)	-	-	-	1 (12.5)	-	1 (16.7)	-	-	-	5 (10.6)	-
Dizziness	1 (25.0)	-	1 (25.0)	-	-	-	-	-	1 (12.5)	-	1 (16.7)	-	1 (33.3)	-	5 (10.6)	-
Myalgia	-	-	1 (25.0)	-	-	-	1 (7.1)	-	-	-	2 (33.3)	-	1 (33.3)	-	5 (10.6)	-
Vomiting	-	-	1 (25.0)	-	1 (12.5)	-	-	-	1 (12.5)	-	1 (16.7)	-	1 (33.3)	-	5 (10.6)	-

*Two Grade 4 events were observed: DL4: CTCL patient with Grade 4 Toxic epidermal necrolysis and an LGL-L patient in DL5 with Grade 4 Neutropenia, both considered not related to KT-333. No Grade 5 events.

Adverse Events Related to KT-333:

- The most common AEs observed in >10% of all patients were stomatitis (38%) and fatigue (17%).
- The only Grade 3 AEs were stomatitis, n=2; arthralgia, n=1; fatigue, n=1; weight decreased, n=1 (there were no >Grade 3 AEs considered related to KT-333).

Serious Adverse Events Related to KT-333:

- Grade 2 pyrexia (n=1) in a patient with NK-Cell lymphoma and Grade 3 stomatitis in a patient with LGL-L (was also a DLT).

Dose Limiting Toxicities:

- Solid tumor/lymphoma patients (n=1): Grade 3 fatigue in a patient treated in DL7.
- Leukemia patients (n=2): Grade 3 stomatitis and Grade 3 arthralgia occurring in two different LGL-L patients treated in DL5.

Exposure, Duration on Treatment and Disposition

- As of 03 June 2024, forty–seven patients received a mean 9.1 doses (range 4.0, 12.3) across the first seven dose levels in patients with solid tumors, lymphomas and LGL-L/T-PLL.
- Seven patients remain active (DL3, n=2; DL6, n=4; DL7, n=1) and 40 patients discontinued KT-333. Primary reasons for discontinuation were disease progression, n=21; discretion of the investigator n=7; adverse event, n=3; withdrawal by patient, n=4; physician decision, n=2, clinical progression, n=2, changes in patient's condition that render them unacceptable for further treatment per PI, n=1.

Pharmacokinetics

Figure 1. Cycle 1, Day 1 Pharmacokinetic Profile and Parameters

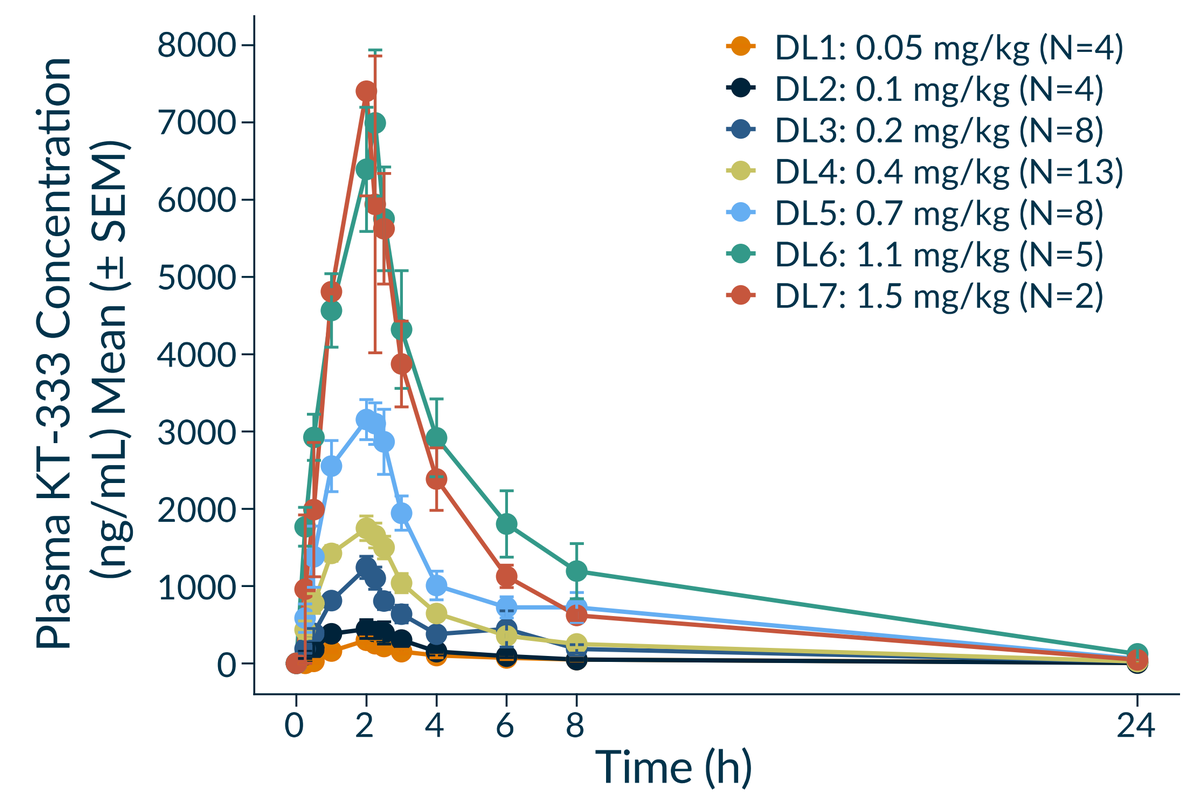
Plasma KT-333 Concentration (ng/mL) Mean (± SEM)
8000
7000
6000
5000
4000
3000
2000
1000
0
0 2 4 6 8 24
Time (h)
DL1: 0.05 mg/kg (N=4)
DL2: 0.1 mg/kg (N=4)
DL3: 0.2 mg/kg (N=8)
DL4: 0.4 mg/kg (N=13)
DL5: 0.7 mg/kg (N=8)
DL6: 1.1 mg/kg (N=5)
DL7: 1.5 mg/kg (N=2)

PK Parameter	0.05 mg/kg (n =4)	0.1 mg/kg (n =3)	0.2 mg/kg (n =8)	0.4 mg/kg (n =13)	0.7 mg/kg (n =8)	1.1 mg/kg (n =4)	1.5 mg/kg (n =2)
C_{max} (ng/mL)	307 (30.5)	443 (24.4)	1330 (37.3)	1880 (25.6)	3580 (24.4)	6380 (27.8)	7410
AUCinf (ng.h/mL)	1550 (66.1)	2060 (24.5)	5990 (64.1)	8610 (44.9)	18800 (36.5)	36500 (54.8)	28000
Vd (L/kg)	0.277 (17.0)	0.289 (17.0)	0.306 (56.5)	0.329 (29.2)	0.316 (52.9)	0.311 (47.4)	0.376
CL (L/h/kg)	0.0448 (62.9)	0.0507 (26.4)	0.0507 (72.3)	0.0554 (43.9)	0.0423 (38.2)	0.0359 (40.4)	0.0539
$T_{1/2}$ (h)	6.25 (78.8)	4.01 (10.1)	4.90 (36.3)	4.42 (21.5)	5.25 (41.2)	6.60 (50.9)	4.72

Mean (%CV) are presented (except where n=2); patient 103-001 (DL2) excluded from calculation of PK parameters due to inconsistency in PK profile

KT-333 exposure increased approximately dose proportionally between the 0.05 mg/kg to 1.5 mg/kg doses, with a half-life of 4-7 hours. Exposure at the 1.5 mg/kg dose was higher than that predicted to be efficacious based on preclinical data.

Clinical Responses

Best Overall Response[1]	Hodgkin's Lymphoma (n=3)	NK-Cell Lymphoma (n=1)	CTCL[3] (n=9)	Solid Tumor (n=14)	Other Hematologic Malignances[4] (n=5)
Complete Response	2	1[2]	-	-	
Partial Response	-	-	4	-	
Stable Disease	1	-	1	4	
Progressive Disease	-	-	4[5]	10	5

[1]The patient totals listed above represent the number of patients enrolled that were disease evaluable for response assessment at the time of cut-off; [2]PET-CR; [3]Cutaneous T-Cell lymphoma; [4]Includes two patients with peripheral T-Cell lymphoma and one each of B-Cell NHL, LGL-L and T-PLL; [5]Includes one patient with clinical progression

Figure 2. Duration of Time on Treatment – Disease Evaluable CTCL, Hodgkin's and NK-Cell Lymphoma Patients

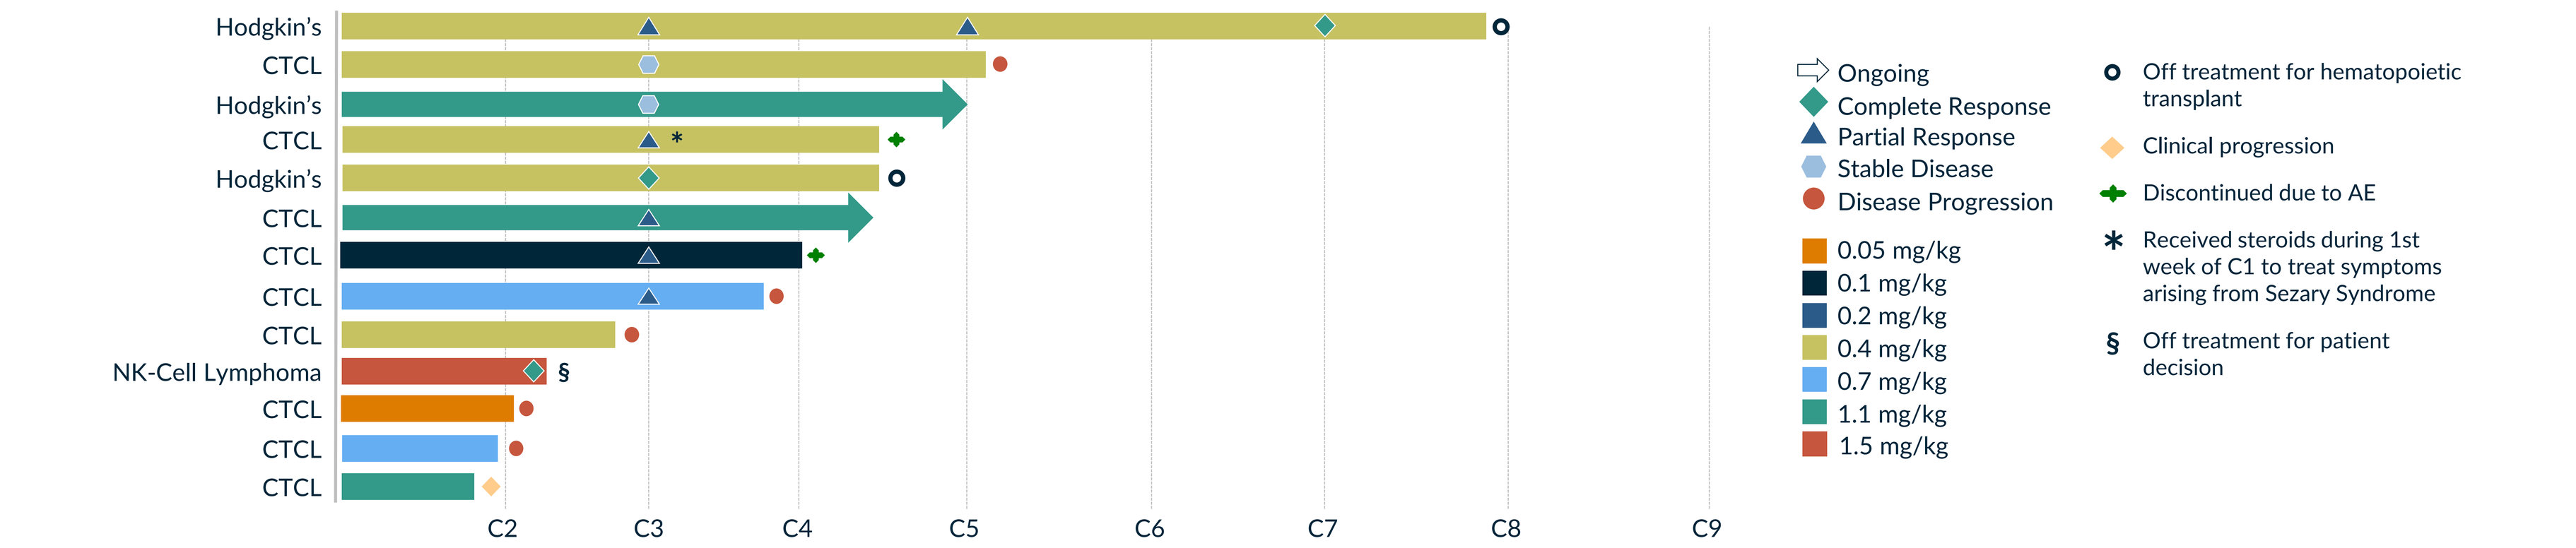
Hodgkin's
CTCL
Hodgkin's
CTCL
Hodgkin's
CTCL
CTCL
CTCL
CTCL
NK-Cell Lymphoma
CTCL
CTCL
CTCL
C2 C3 C4 C5 C6 C7 C8 C9
Ongoing
Complete Response
Partial Response
Stable Disease
Disease Progression
0.05 mg/kg
0.1 mg/kg
0.2 mg/kg
0.4 mg/kg
0.7 mg/kg
1.1 mg/kg
1.5 mg/kg
Off treatment for hematopoietic transplant
Clinical progression
Discontinued due to AE
Received steroids during 1st week of C1 to treat symptoms arising from Sezary Syndrome
§ Off treatment for patient decision

Pharmacodynamics

Figure 3. KT-333 leads to Mean Maximum STAT3 Degradation of Up to 95% in Peripheral Blood Mononuclear Cells at Dose Level 7 Demonstrating Proof-of-Mechanism

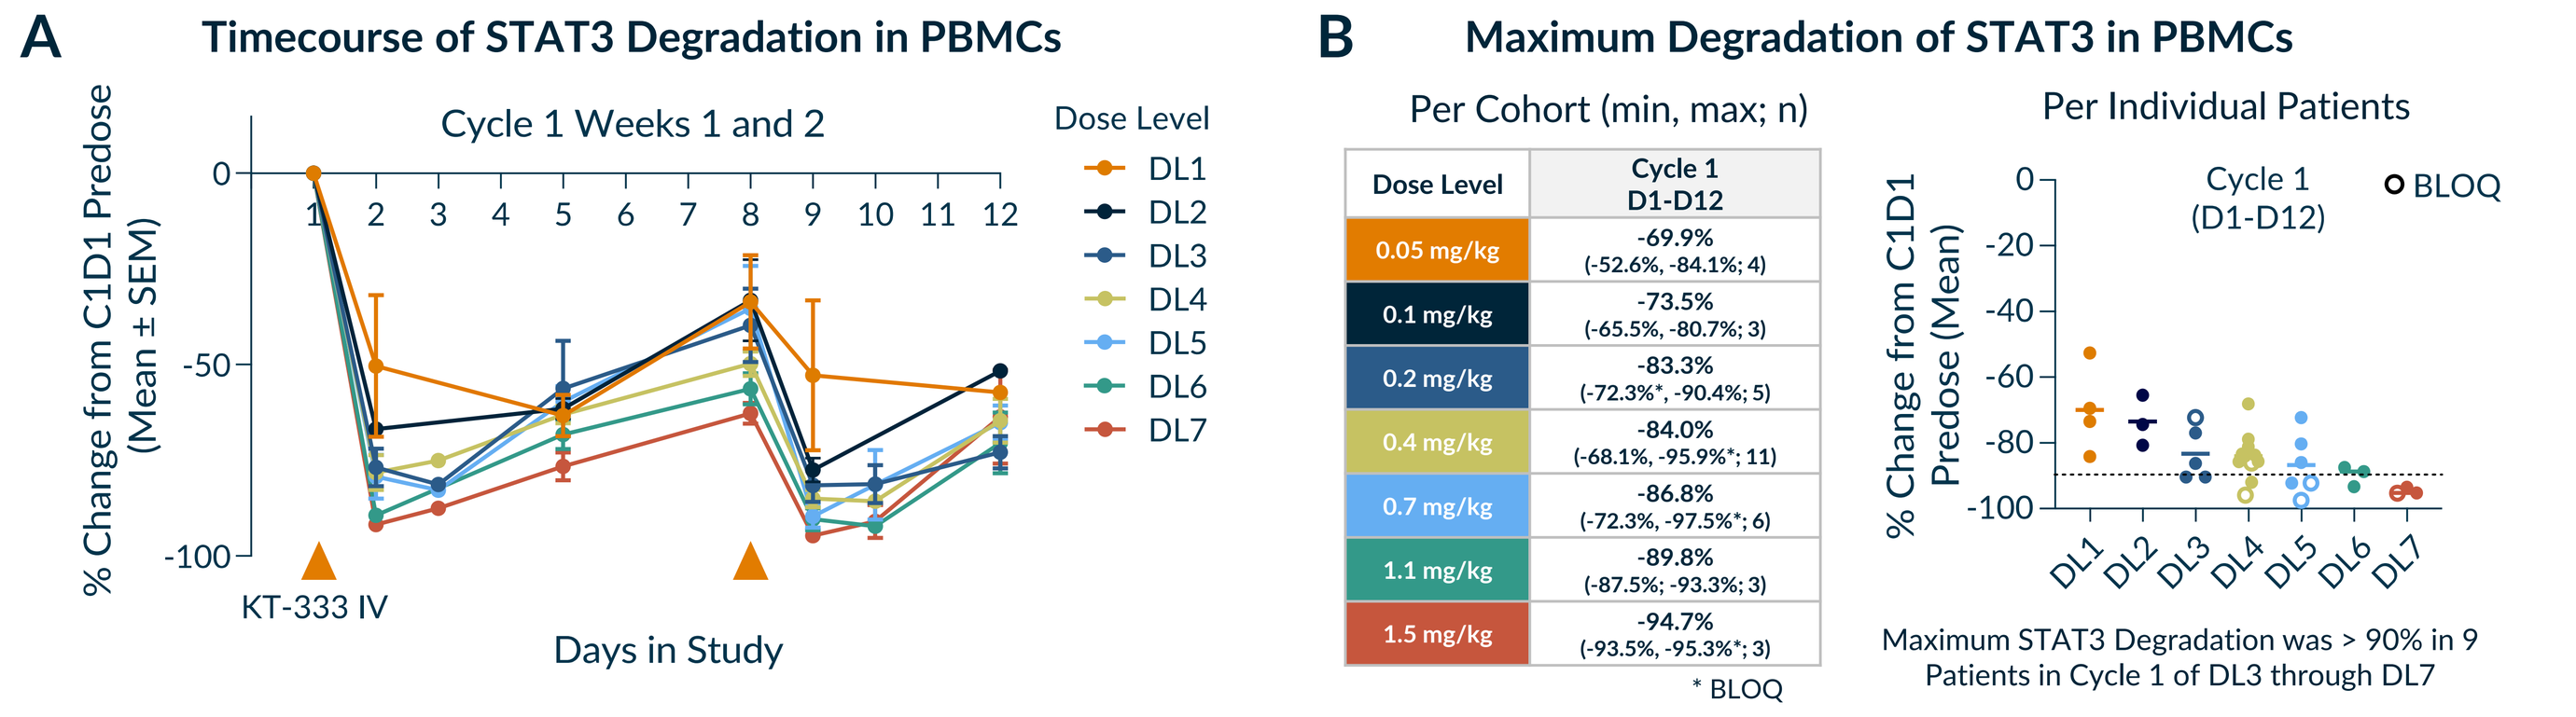
A
Timecourse of STAT3 Degradation in PBMCs
Cycle 1 Weeks 1 and 2
% Change from C1D1 Predose (Mean ± SEM)
0
-50
-100
1 2 3 4 5 6 7 8 9 10 11 12
KT-333 IV
Days in Study
Dose Level
DL1
DL2
DL3
DL4
DL5
DL6
DL7
B
Maximum Degradation of STAT3 in PBMCs
Per Cohort (min, max; n)
Dose Level | Cycle 1 D1-D12
0.05 mg/kg | -69.9% (-52.6%, -84.1%; 4)
0.1 mg/kg | -73.5% (-65.5%, -80.7%; 3)
0.2 mg/kg | -83.3% (-72.3%*, -90.4%; 5)
0.4 mg/kg | -84.0% (-68.1%, -95.9%*; 11)
0.7 mg/kg | -86.8% (-72.3%, -97.5%*; 6)
1.1 mg/kg | -89.8% (-87.5%, -93.3%; 3)
1.5 mg/kg | -94.7% (-93.5%, -95.3%*; 3)
* BLOQ
Per Individual Patients
% Change from C1D1 Predose (Mean)
Cycle 1 (D1-D12)
BLOQ
0
-20
-40
-60
-80
-100
DL1 DL2 DL3 DL4 DL5 DL6 DL7
Maximum STAT3 Degradation was > 90% in 9 Patients in Cycle 1 of DL3 through DL7

A, B) Percent change in STAT3 represents mean percent change of two STAT3 peptides from baseline measured using targeted mass spectrometry (MS) assay. Screening sample was used as baseline when C1D1 predose was not available. When both samples were available, C1D1 predose data served as baseline. For measurements BLOQ, 50% of LLOQ values for the respective STAT3 peptides were used for computation of change from baseline. DL5 data includes PD after dose reductions in two LGL-L patients (i) on C1D22 for one patient and (ii) on C1D8 and C2D1 for the other patient, both due to AEs.

Figure 4. KT-333 Leads to Marked Reductions in STAT3, pSTAT3 and SOCS3 Levels with Concomitant Induction of IFNγ Stimulated Genes Including Chemokines, CXCL9 and CXCL10 in Tumor Tissue from a CTCL Patient in DL4

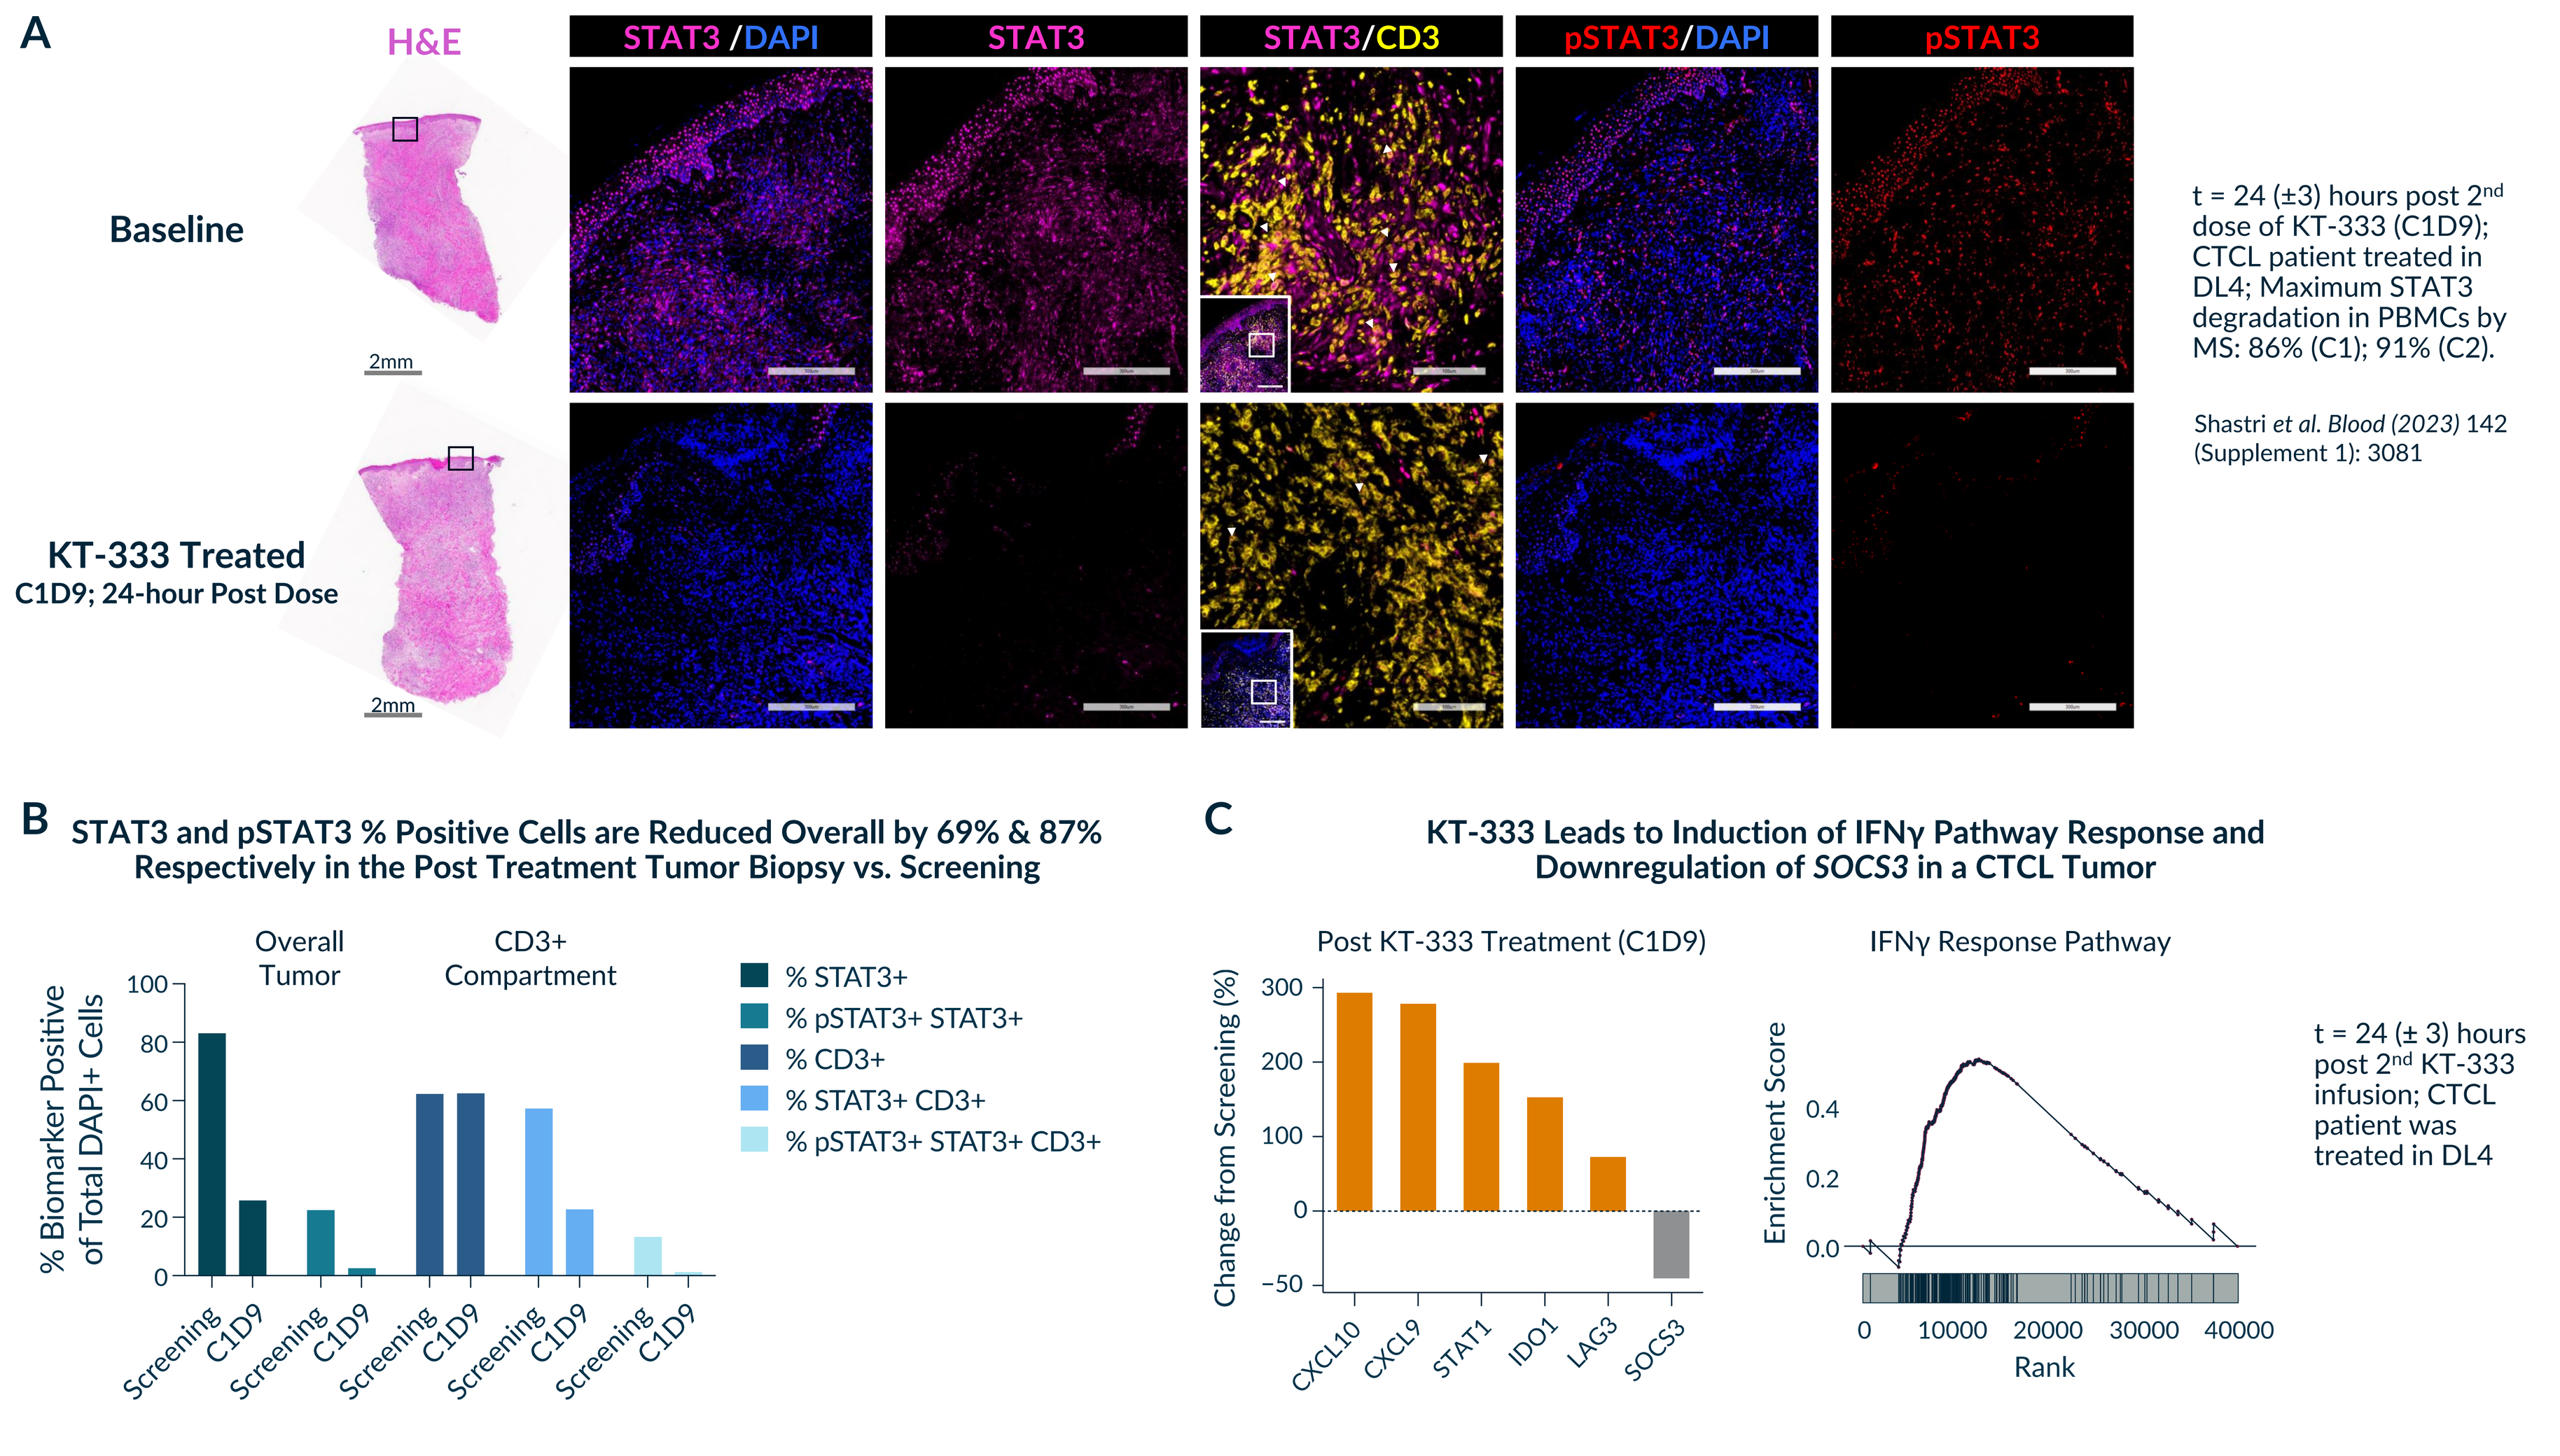
A
H&E
STAT3 /DAPI
STAT3
STAT3/CD3
pSTAT3/DAPI
pSTAT3
Baseline
KT-333 Treated
C1D9; 24-hour Post Dose
2mm
2mm
t = 24 (±3) hours post 2nd dose of KT-333 (C1D9); CTCL patient treated in DL4; Maximum STAT3 degradation in PBMCs by MS: 86% (C1); 91% (C2).
Shastri et al. Blood (2023) 142 (Supplement 1): 3081
B
STAT3 and pSTAT3 % Positive Cells are Reduced Overall by 69% & 87% Respectively in the Post Treatment Tumor Biopsy vs. Screening
Overall Tumor
CD3+ Compartment
% Biomarker Positive of Total DAPI+ Cells
100
80
60
40
20
0
Screening C1D9 Screening C1D9 Screening C1D9 Screening C1D9 Screening C1D9
% STAT3+
% pSTAT3+ STAT3+
% CD3+
% STAT3+ CD3+
% pSTAT3+ STAT3+ CD3+
C
KT-333 Leads to Induction of IFNγ Pathway Response and Downregulation of SOCS3 in a CTCL Tumor
Post KT-333 Treatment (C1D9)
Change from Screening (%)
300
200
100
0
-50
CXCL10 CXCL9 STAT1 IDO1 LAG3 SOCS3
IFNγ Response Pathway
Enrichment Score
0.4
0.2
0.0
0 10000 20000 30000 40000
Rank
t = 24 (± 3) hours post 2nd KT-333 infusion; CTCL patient was treated in DL4

A) Representative ROIs from histological sections of tumor biopsies analyzed using multiplex immunofluorescence (mIF) for STAT3, pSTAT3 and CD3. Images include epidermal region, dermal region and CD3+ tumor infiltrate. DAPI was used as nuclear counterstain. Scale bars: 2 mm for H&E; 300 µm for mIF and 80 µm for high magnification ROI showing STAT3/CD3 co-stain. White arrows indicate STAT3+CD3+ cells. **B**) HALO image analysis platform was used for analysis of mIF data. Intensity-based thresholds were set to derive classifiers for CD3, STAT3, pSTAT3 that delineated biomarker positive versus negative DAPI+ cells in both the screening and C1D9 biopsies. The epidermis was excluded from quantitative analysis. **C**) Gene expression profiling of FFPE specimen from screening and C1D9 CTCL biopsy using RNA sequencing.

CONCLUSIONS

- KT-333 was well tolerated with primarily Grade 1 and 2 adverse events. Two DLTs occurred in LGL-L patients at DL5 and one DLT was observed in a lymphoma patient treated at DL7. Dose escalation is ongoing at DL7 in solid tumor/lymphoma patients and complete at DL4 in leukemia patients.
- Complete response (CR) achieved in 2 of 3 patients with Hodgkin's lymphoma at DL4 who both received prior brentuximab vedotin and at least one regimen containing a check point inhibitor, and partial response (PR) was achieved in 4 of 9 evaluable CTCL patients treated at DL2 and DL4-6. CR observed in NK-Cell Lymphoma patient with STAT3 mutation at DL7.
- KT-333 achieved up to 95% mean maximum STAT3 degradation in peripheral blood mononuclear cells at DL7.
- KT-333 resulted in substantial reduction of STAT3, pSTAT3 and SOCS3 in a CTCL patient tumor with concomitant induction of IFNγ-stimulated genes, including chemokines CXCL9 and CXCL10, suggestive of a favorable immunomodulatory response in the TME.
- Results overall show robust STAT3 knockdown and favorable immunomodulation in blood and tumor at doses that were well-tolerated and predicted to be clinically efficacious, with promising antitumor activity in heavily pre-treated cHL, CTCL and STAT3mut NK-Cell Lymphoma patients.

ACKNOWLEDGMENT

Kymera Therapeutics would like to thank the patients for their participation in the clinical study and their family and friends as well as the support of the clinical sites.

KYMERA Kymera Therapeutics, 500 North Beacon, 4th Floor, Watertown, MA 02472 www.kymeratx.com

The safety and efficacy of this investigational agent have not been established. This agent is not approved by any regulatory body for any indication.

European Hematology Association • June 13-16, 2024 • Madrid Spain